Spatiotemporal Context Shapes Center-Surround Dynamics

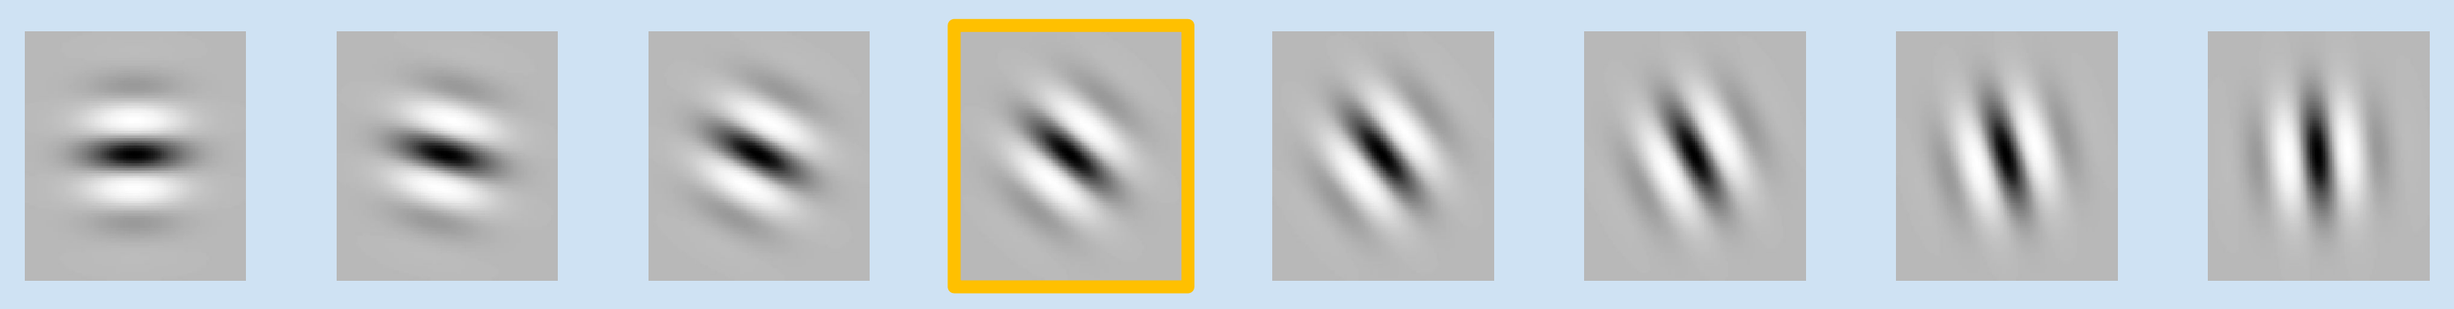

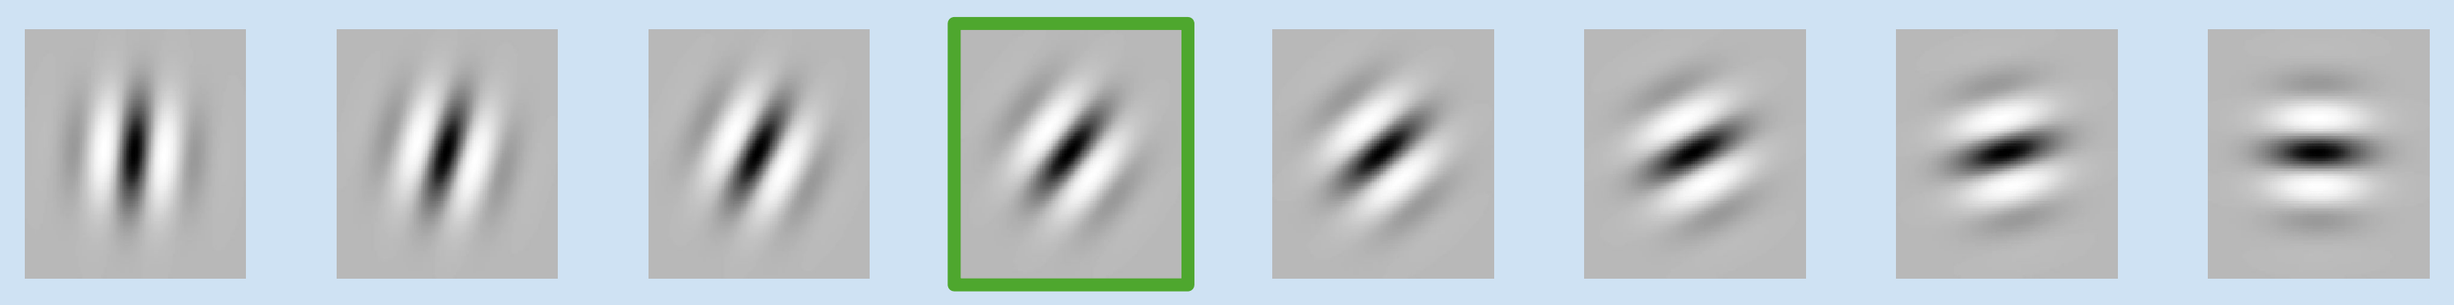

Lisa Schwetlick[1,2]

Peter Neri[2]

[1] Laboratory of Psychophysics, EPFL, Lausanne, Switzerland

[2] Laboratory of Perceptice Systems, ENS, Paris, France

Correspondence: lisa.schwetlick@epfl.ch

Method

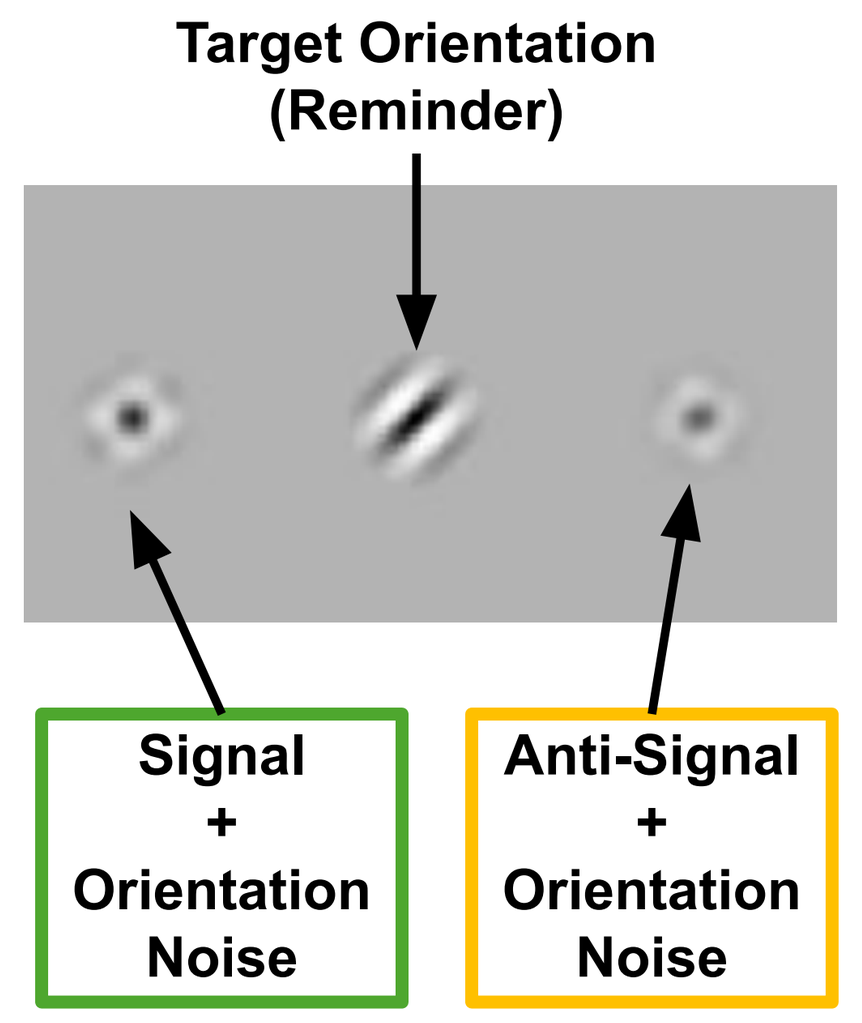

Target Orientation (Reminder)
Signal + Orientation Noise
Anti-Signal + Orientation Noise

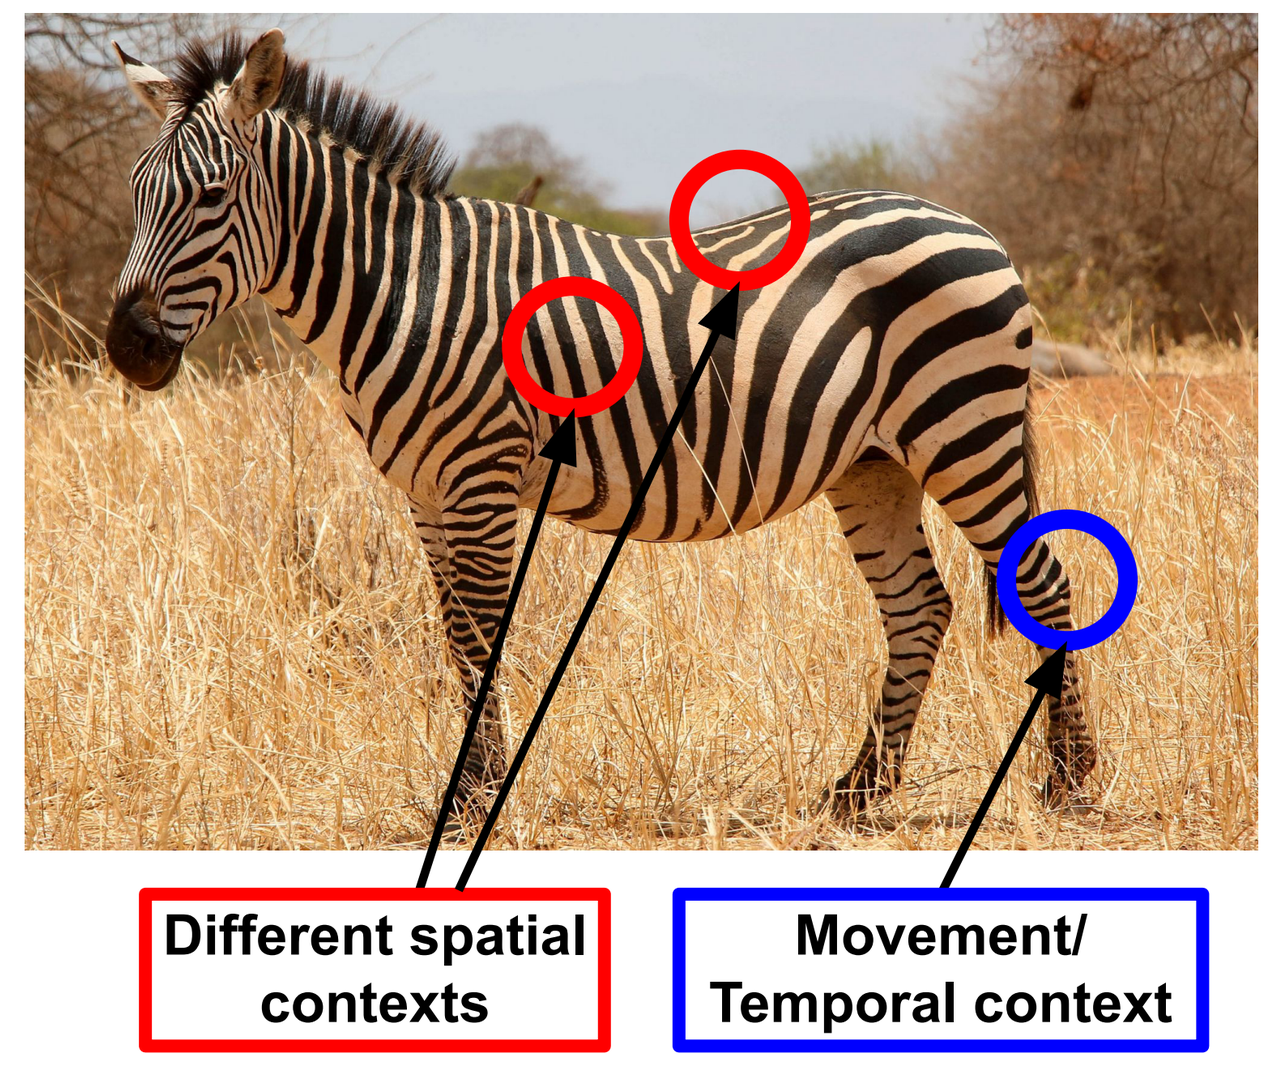

Different spatial contexts
Movement/ Temporal context

- **RQ:** In nature, stimuli exist in context, both in time and in space. How does the judgment of orientation interact with the context?
- **Task**: "is the target orientation on the left or right?"
- No **eye movement** (fixation marker)
- Presentation time **80ms**
- 16 **Participants** (so far)
- **Trials**: ~75 000
- Eccentricity: 4°

Reverse Correlation

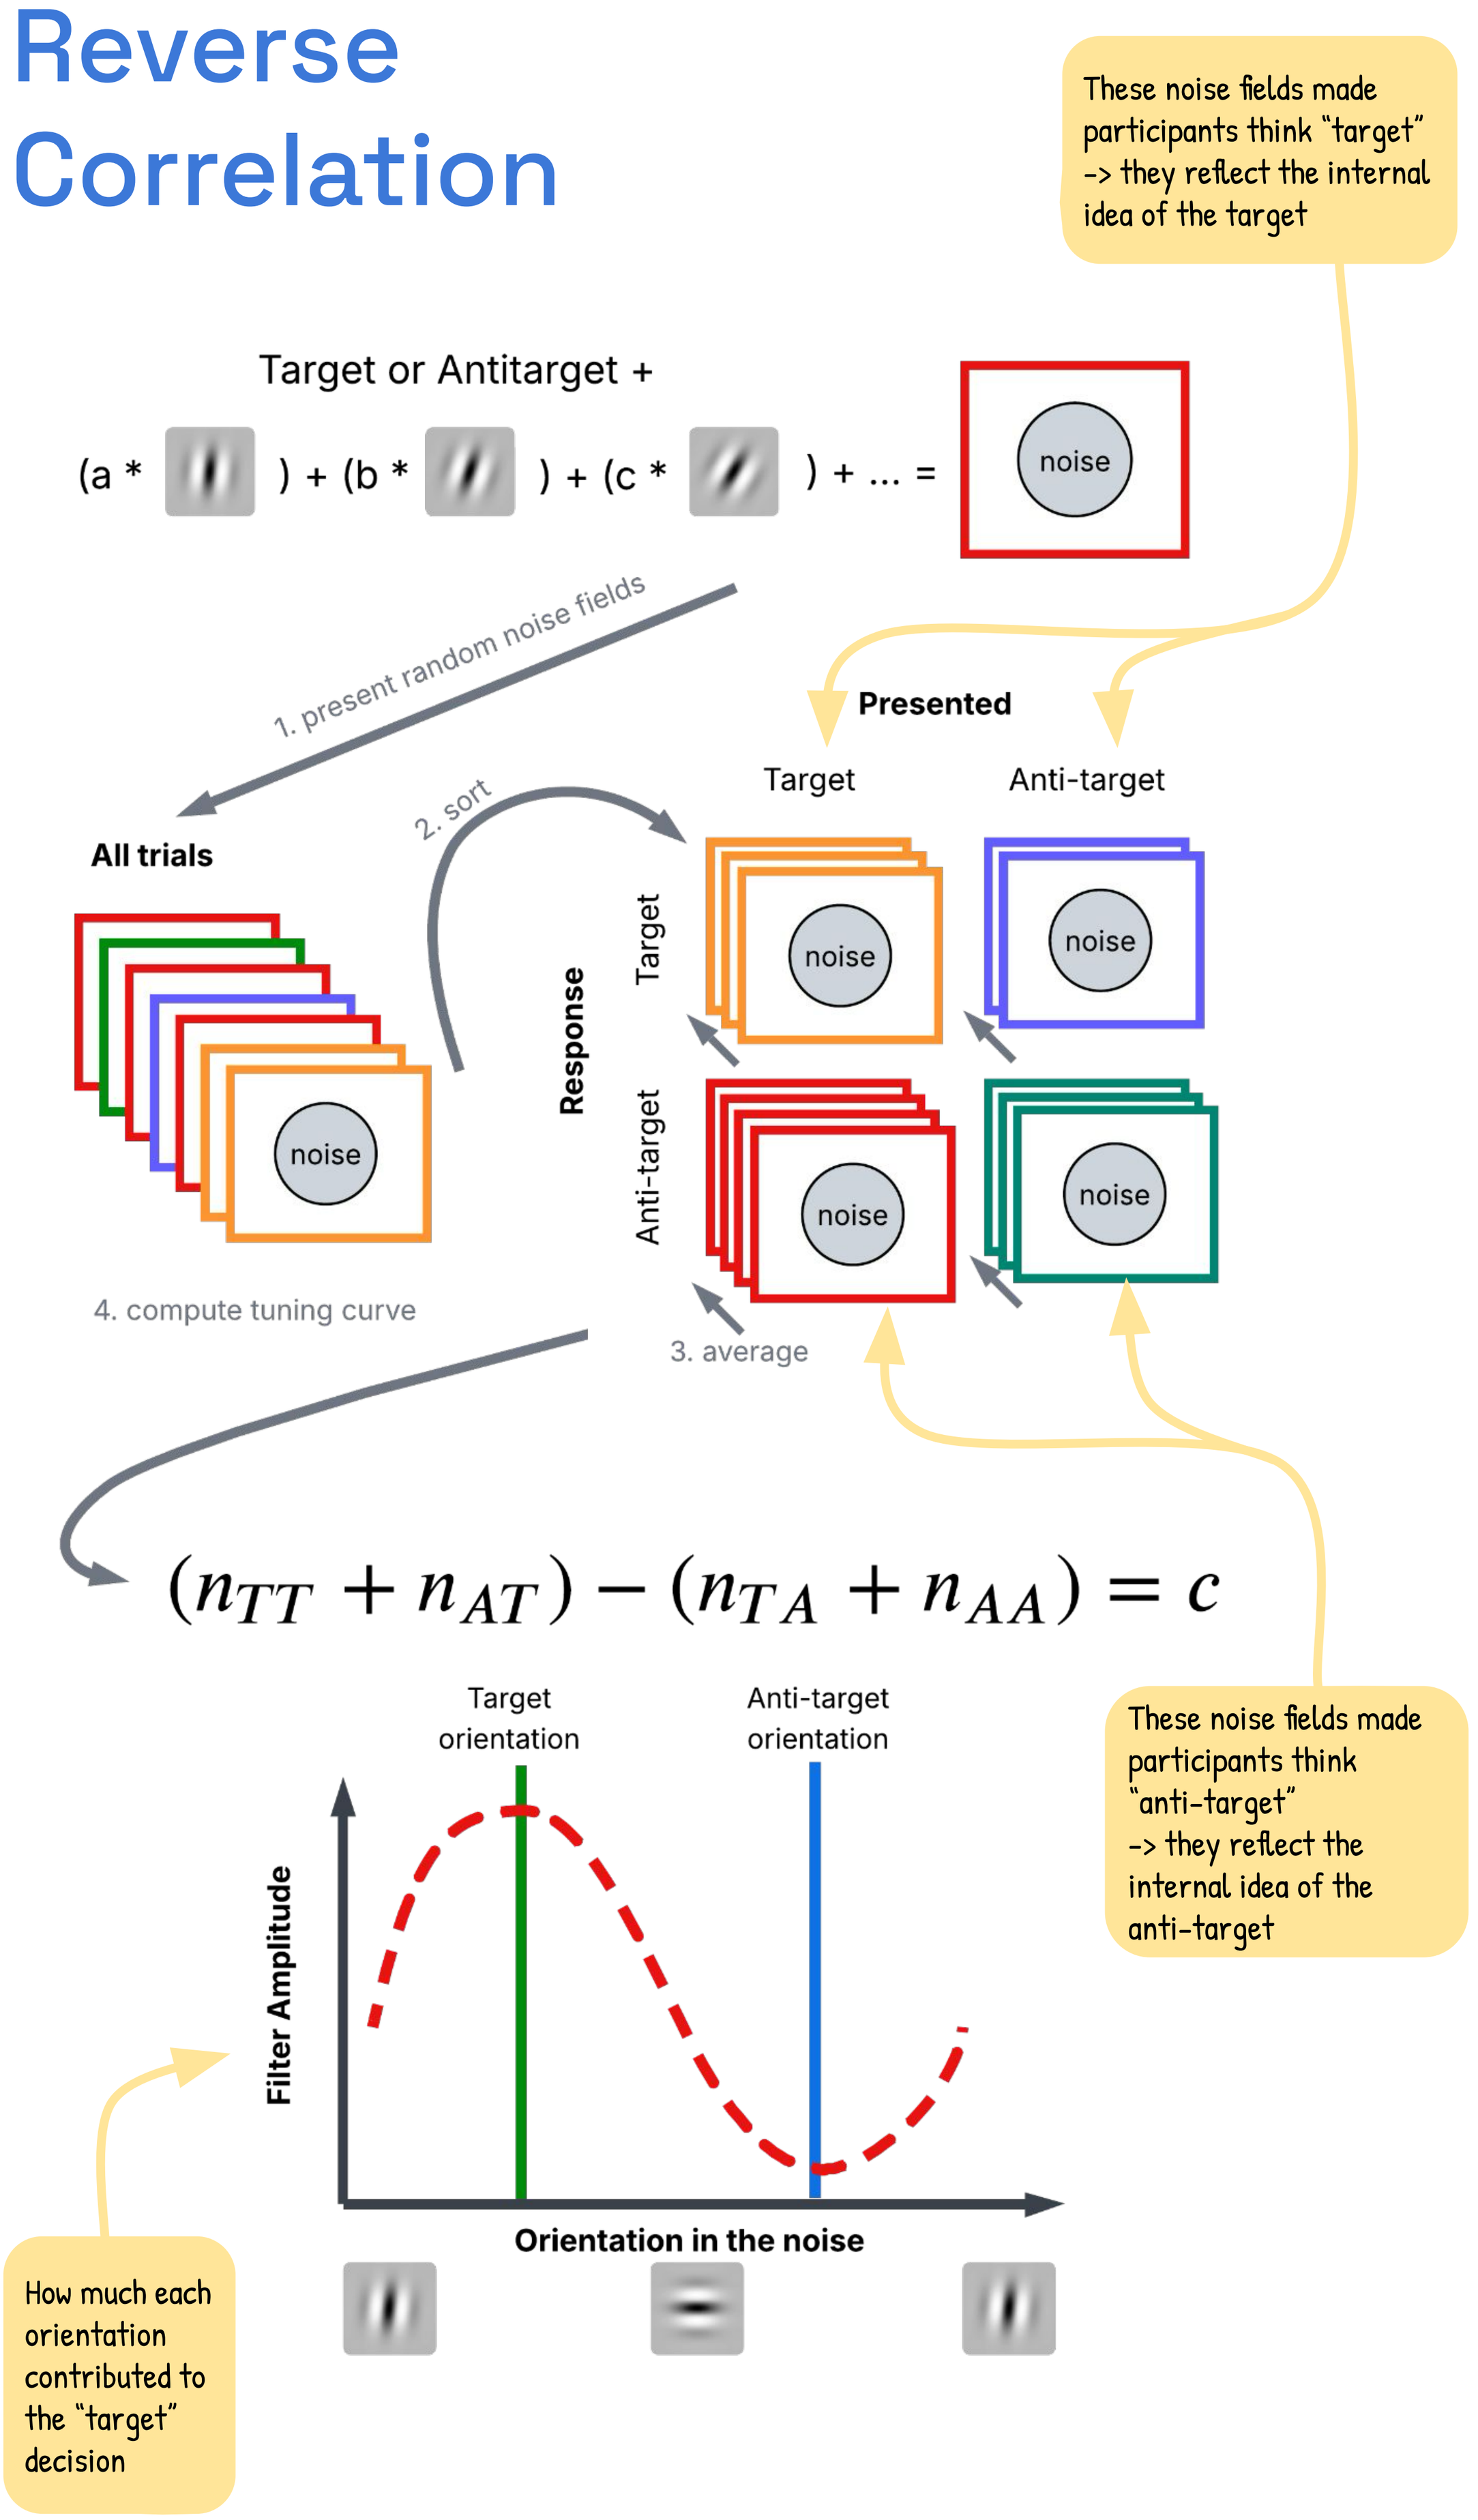

These noise fields made participants think "target" -> they reflect the internal idea of the target
Target or Antitarget +
(a *) + (b *) + (c *) + ... =
noise
1. present random noise fields
2. sort
Presented
Target
Anti-target
All trials
Response
Target
Anti-target
4. compute tuning curve
3. average
$(n_{TT} + n_{AT}) - (n_{TA} + n_{AA}) = c$
Target orientation
Anti-target orientation
These noise fields made participants think "anti-target" -> they reflect the internal idea of the anti-target
Filter Amplitude
Orientation in the noise
How much each orientation contributed to the "target" decision

Spatial Context

Stimuli

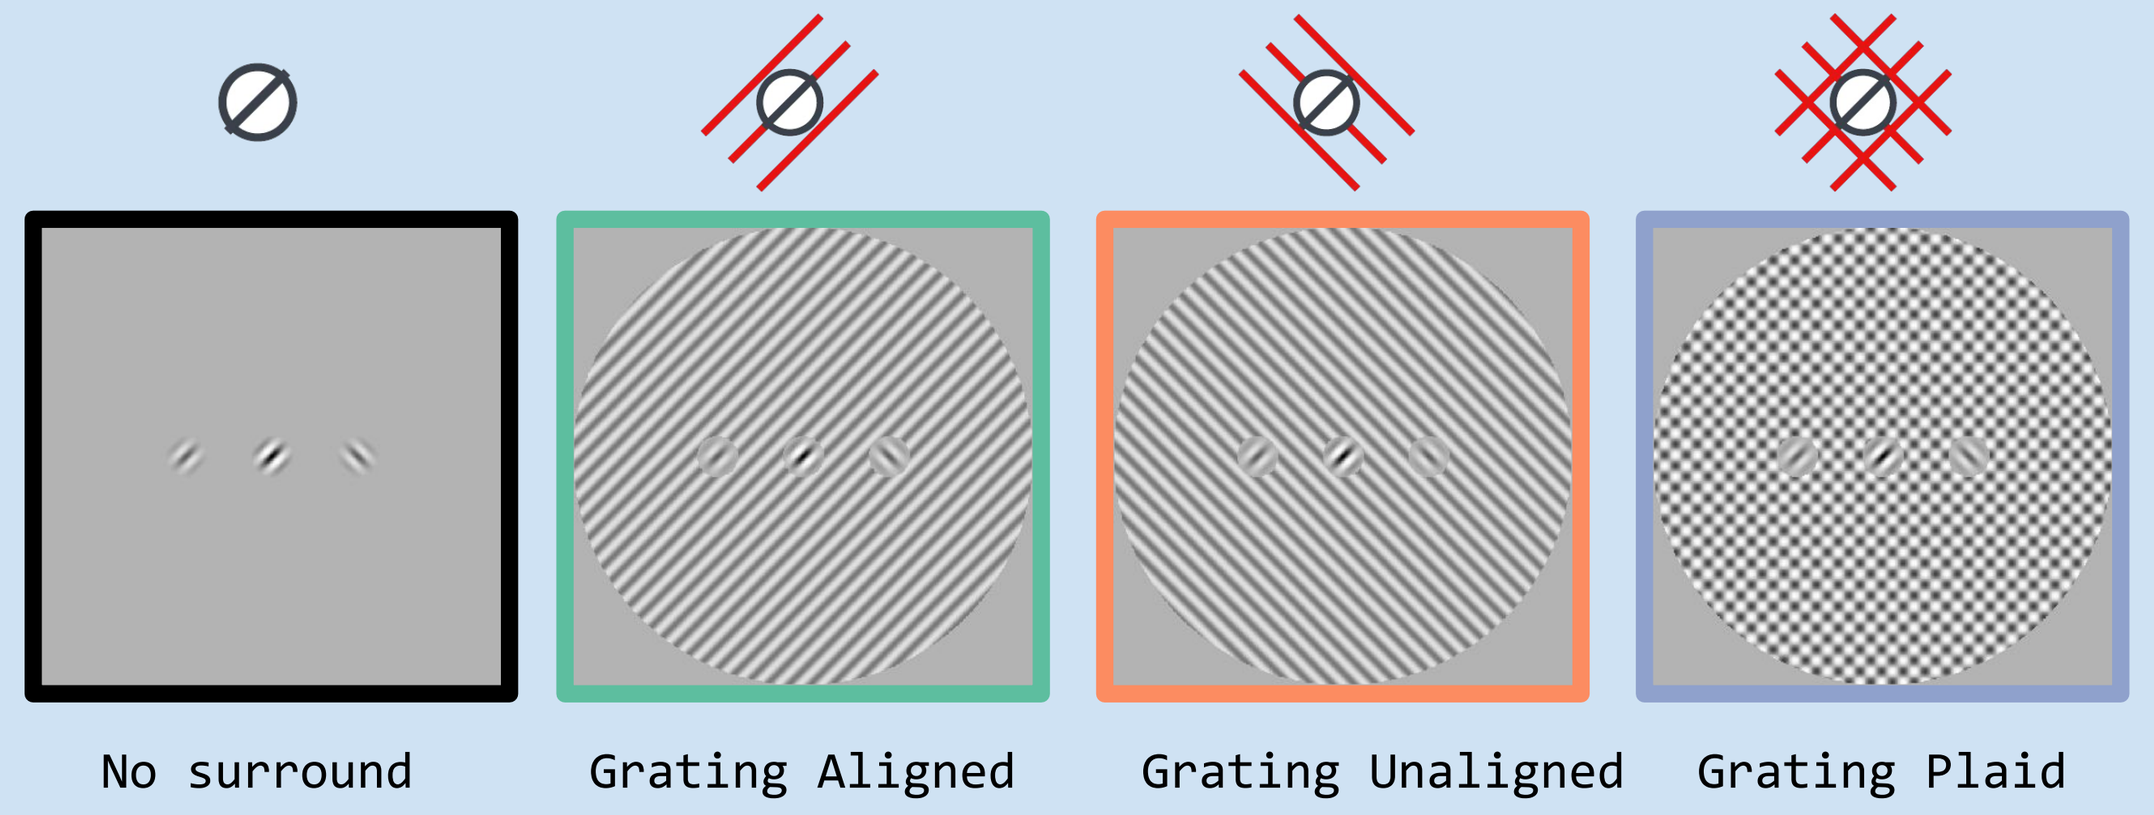

No surround
Grating Aligned
Grating Unaligned
Grating Plaid

Results

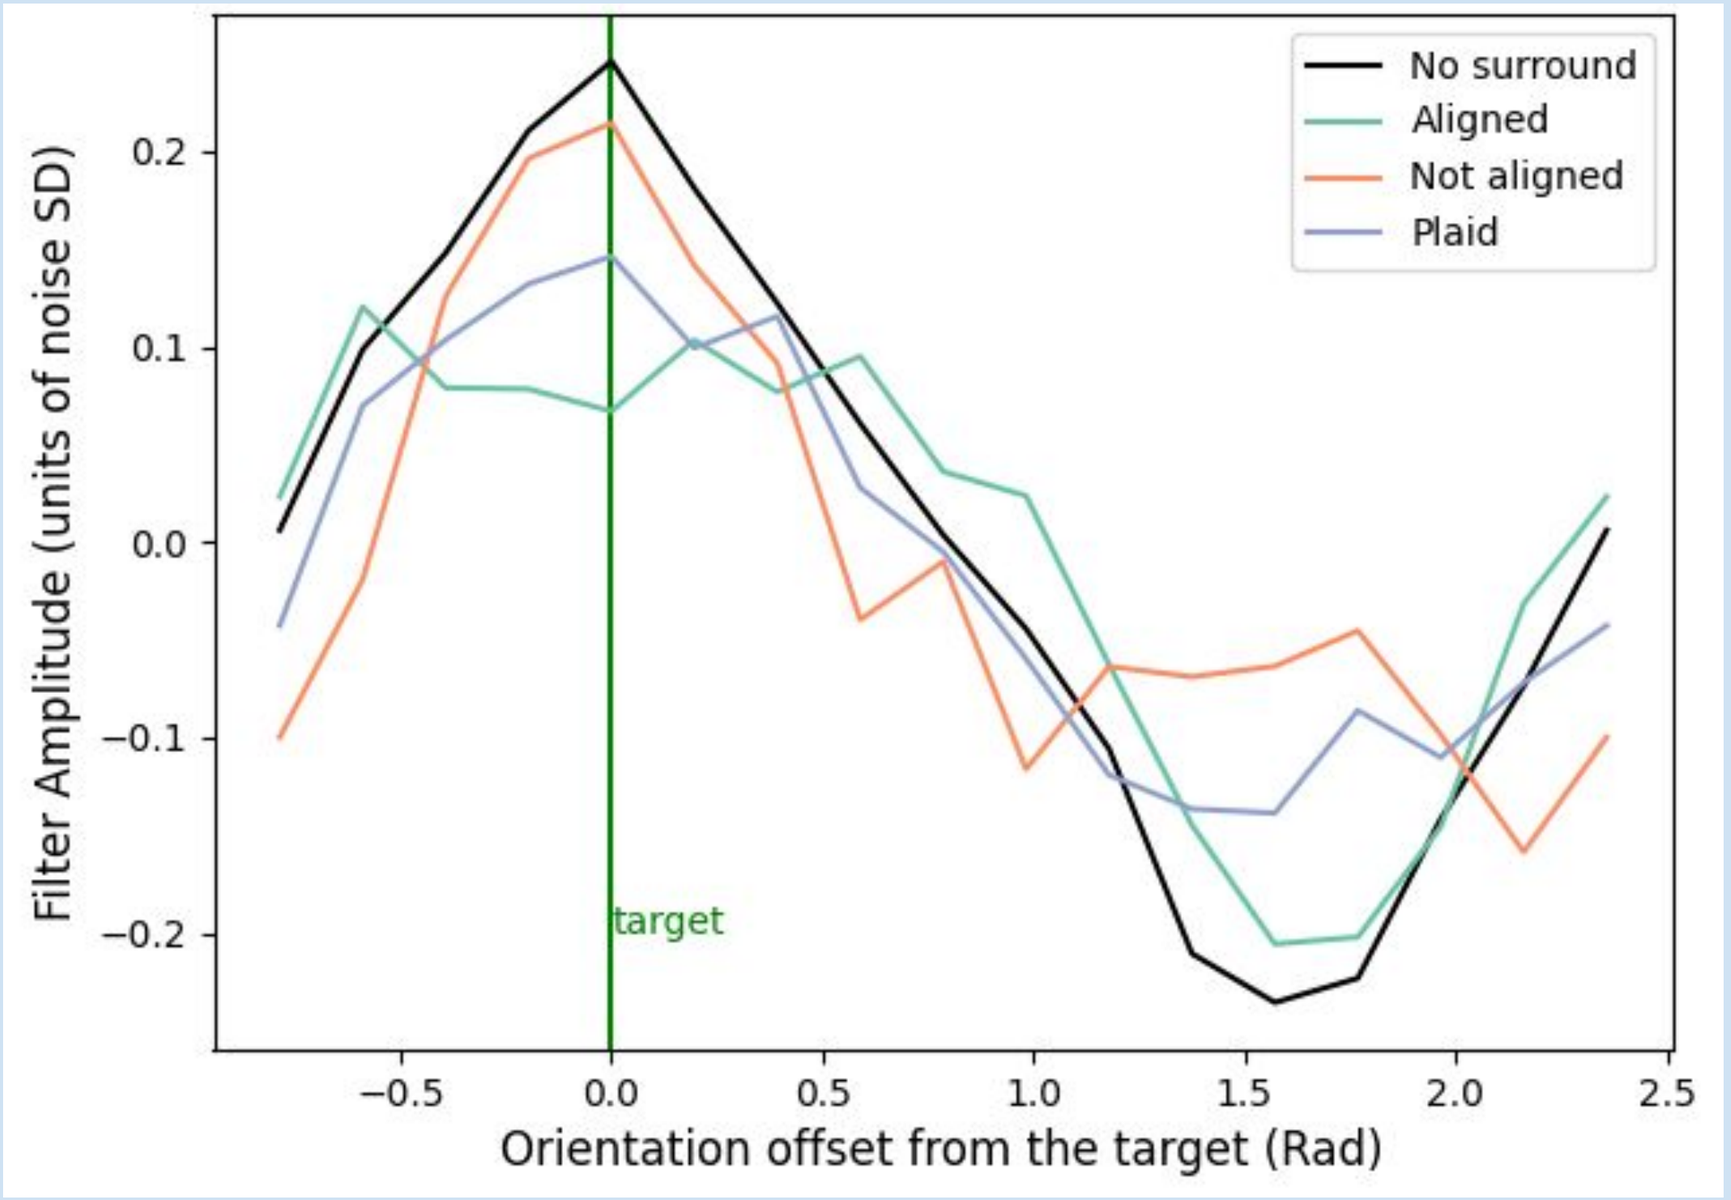

No surround
Aligned
Not aligned
Plaid
Filter Amplitude (units of noise SD)
target
Orientation offset from the target (Rad)

So what?

Target-aligned surrounds attenuate the tuning curve selectively at the target orientation. **Anti-target-aligned surrounds** attenuate the tuning curve selectively at the anti-target orientation.

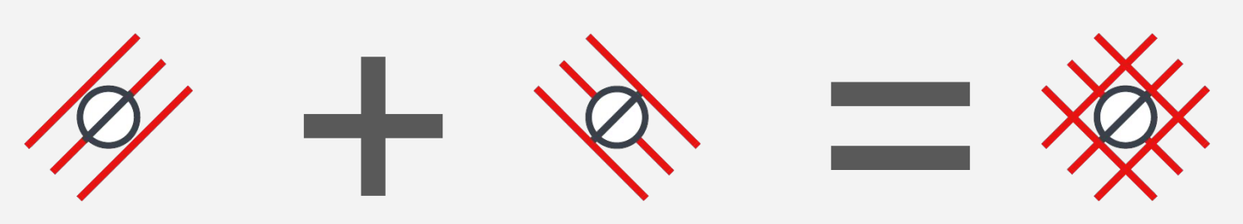

Individuals

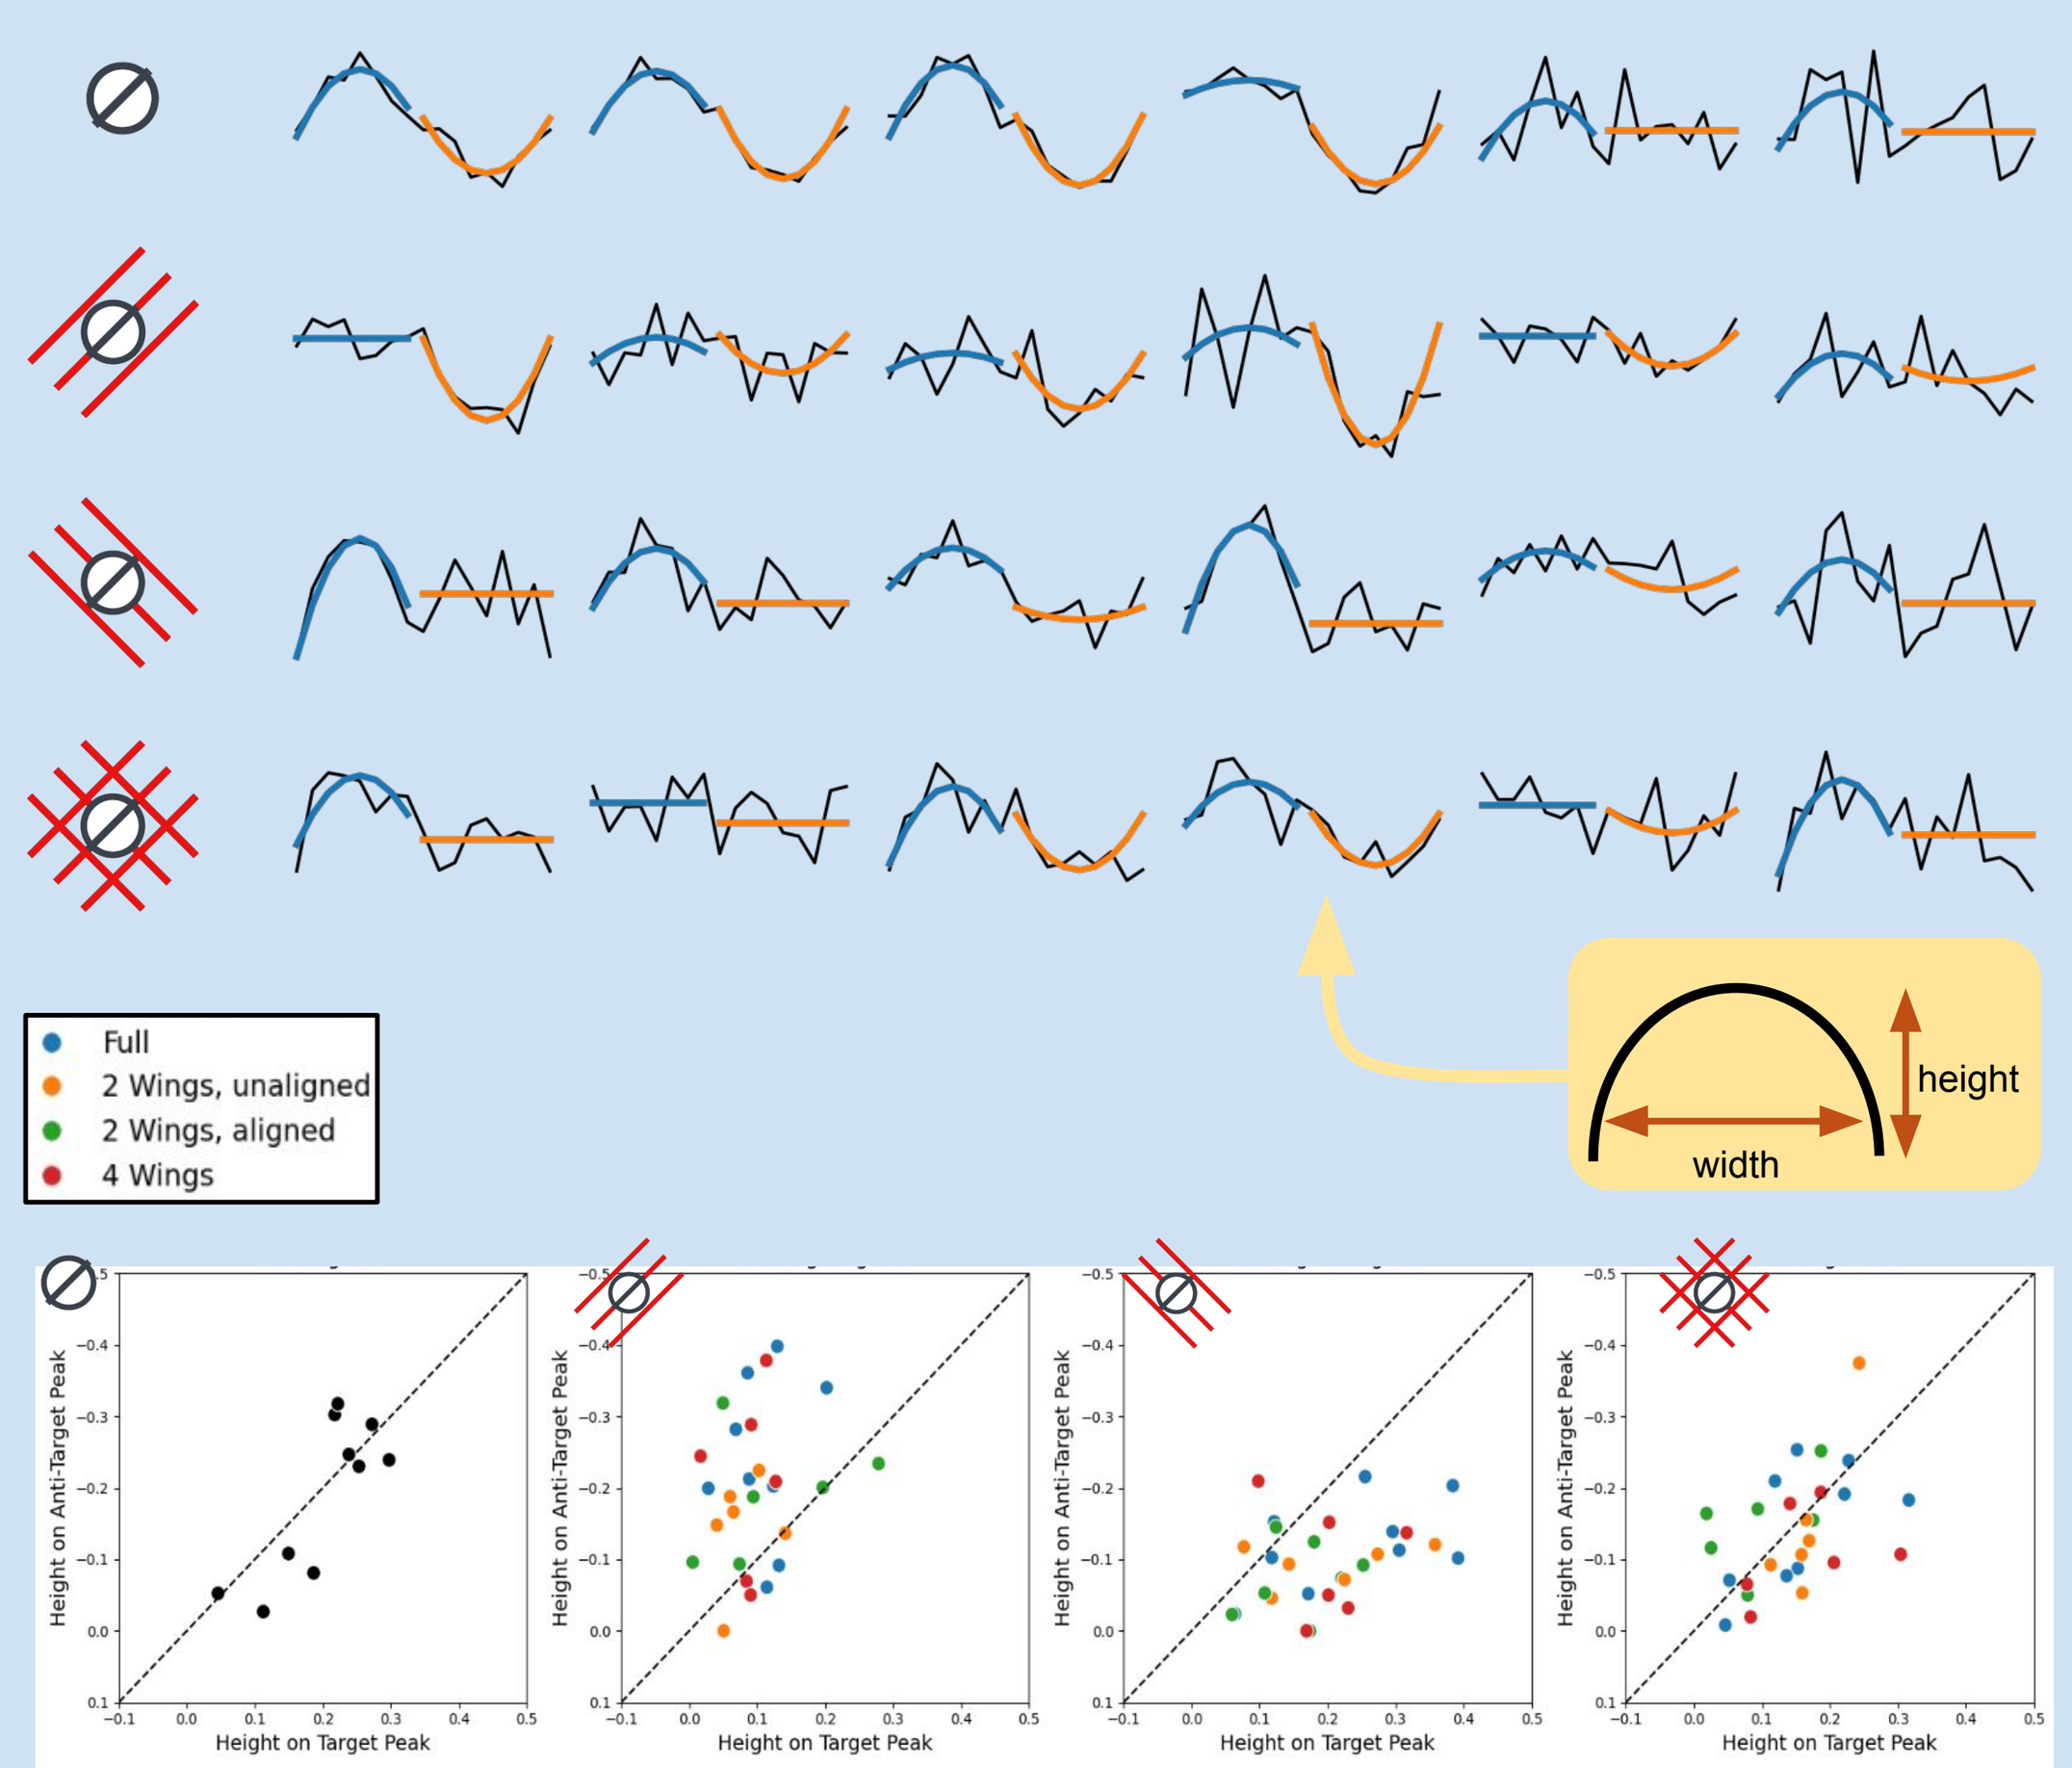

participants
Full
2 Wings, unaligned
2 Wings, aligned
4 Wings
height
width
Height on Anti-Target Peak
Height on Target Peak

Global Spatial Context

Stimuli

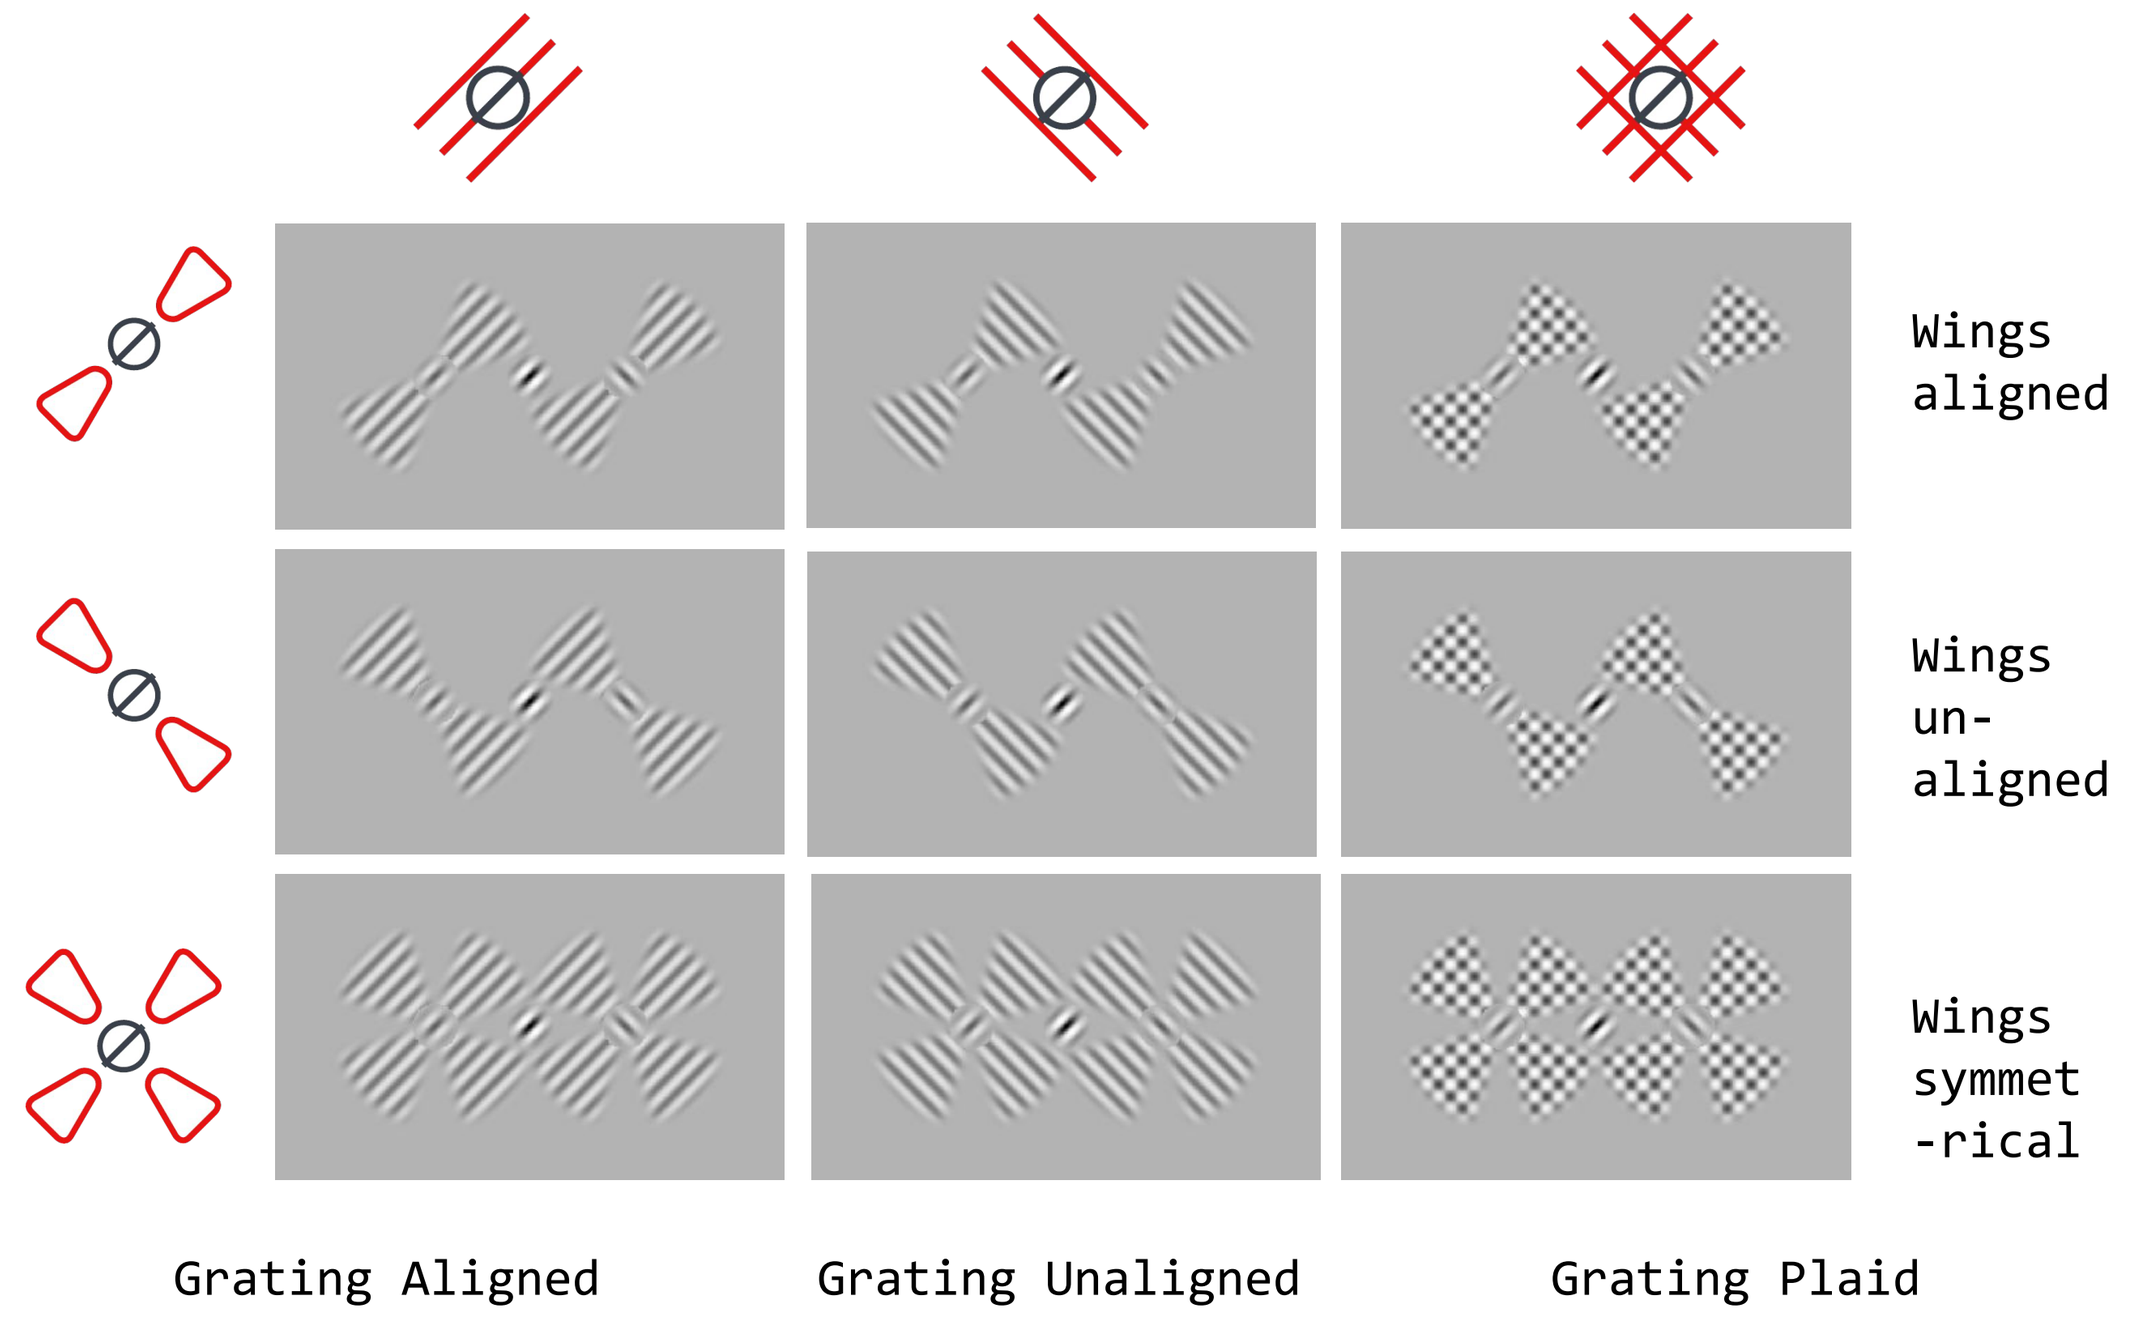

Wings aligned
Wings un-aligned
Wings symmet-rical
Grating Aligned
Grating Unaligned
Grating Plaid

Results

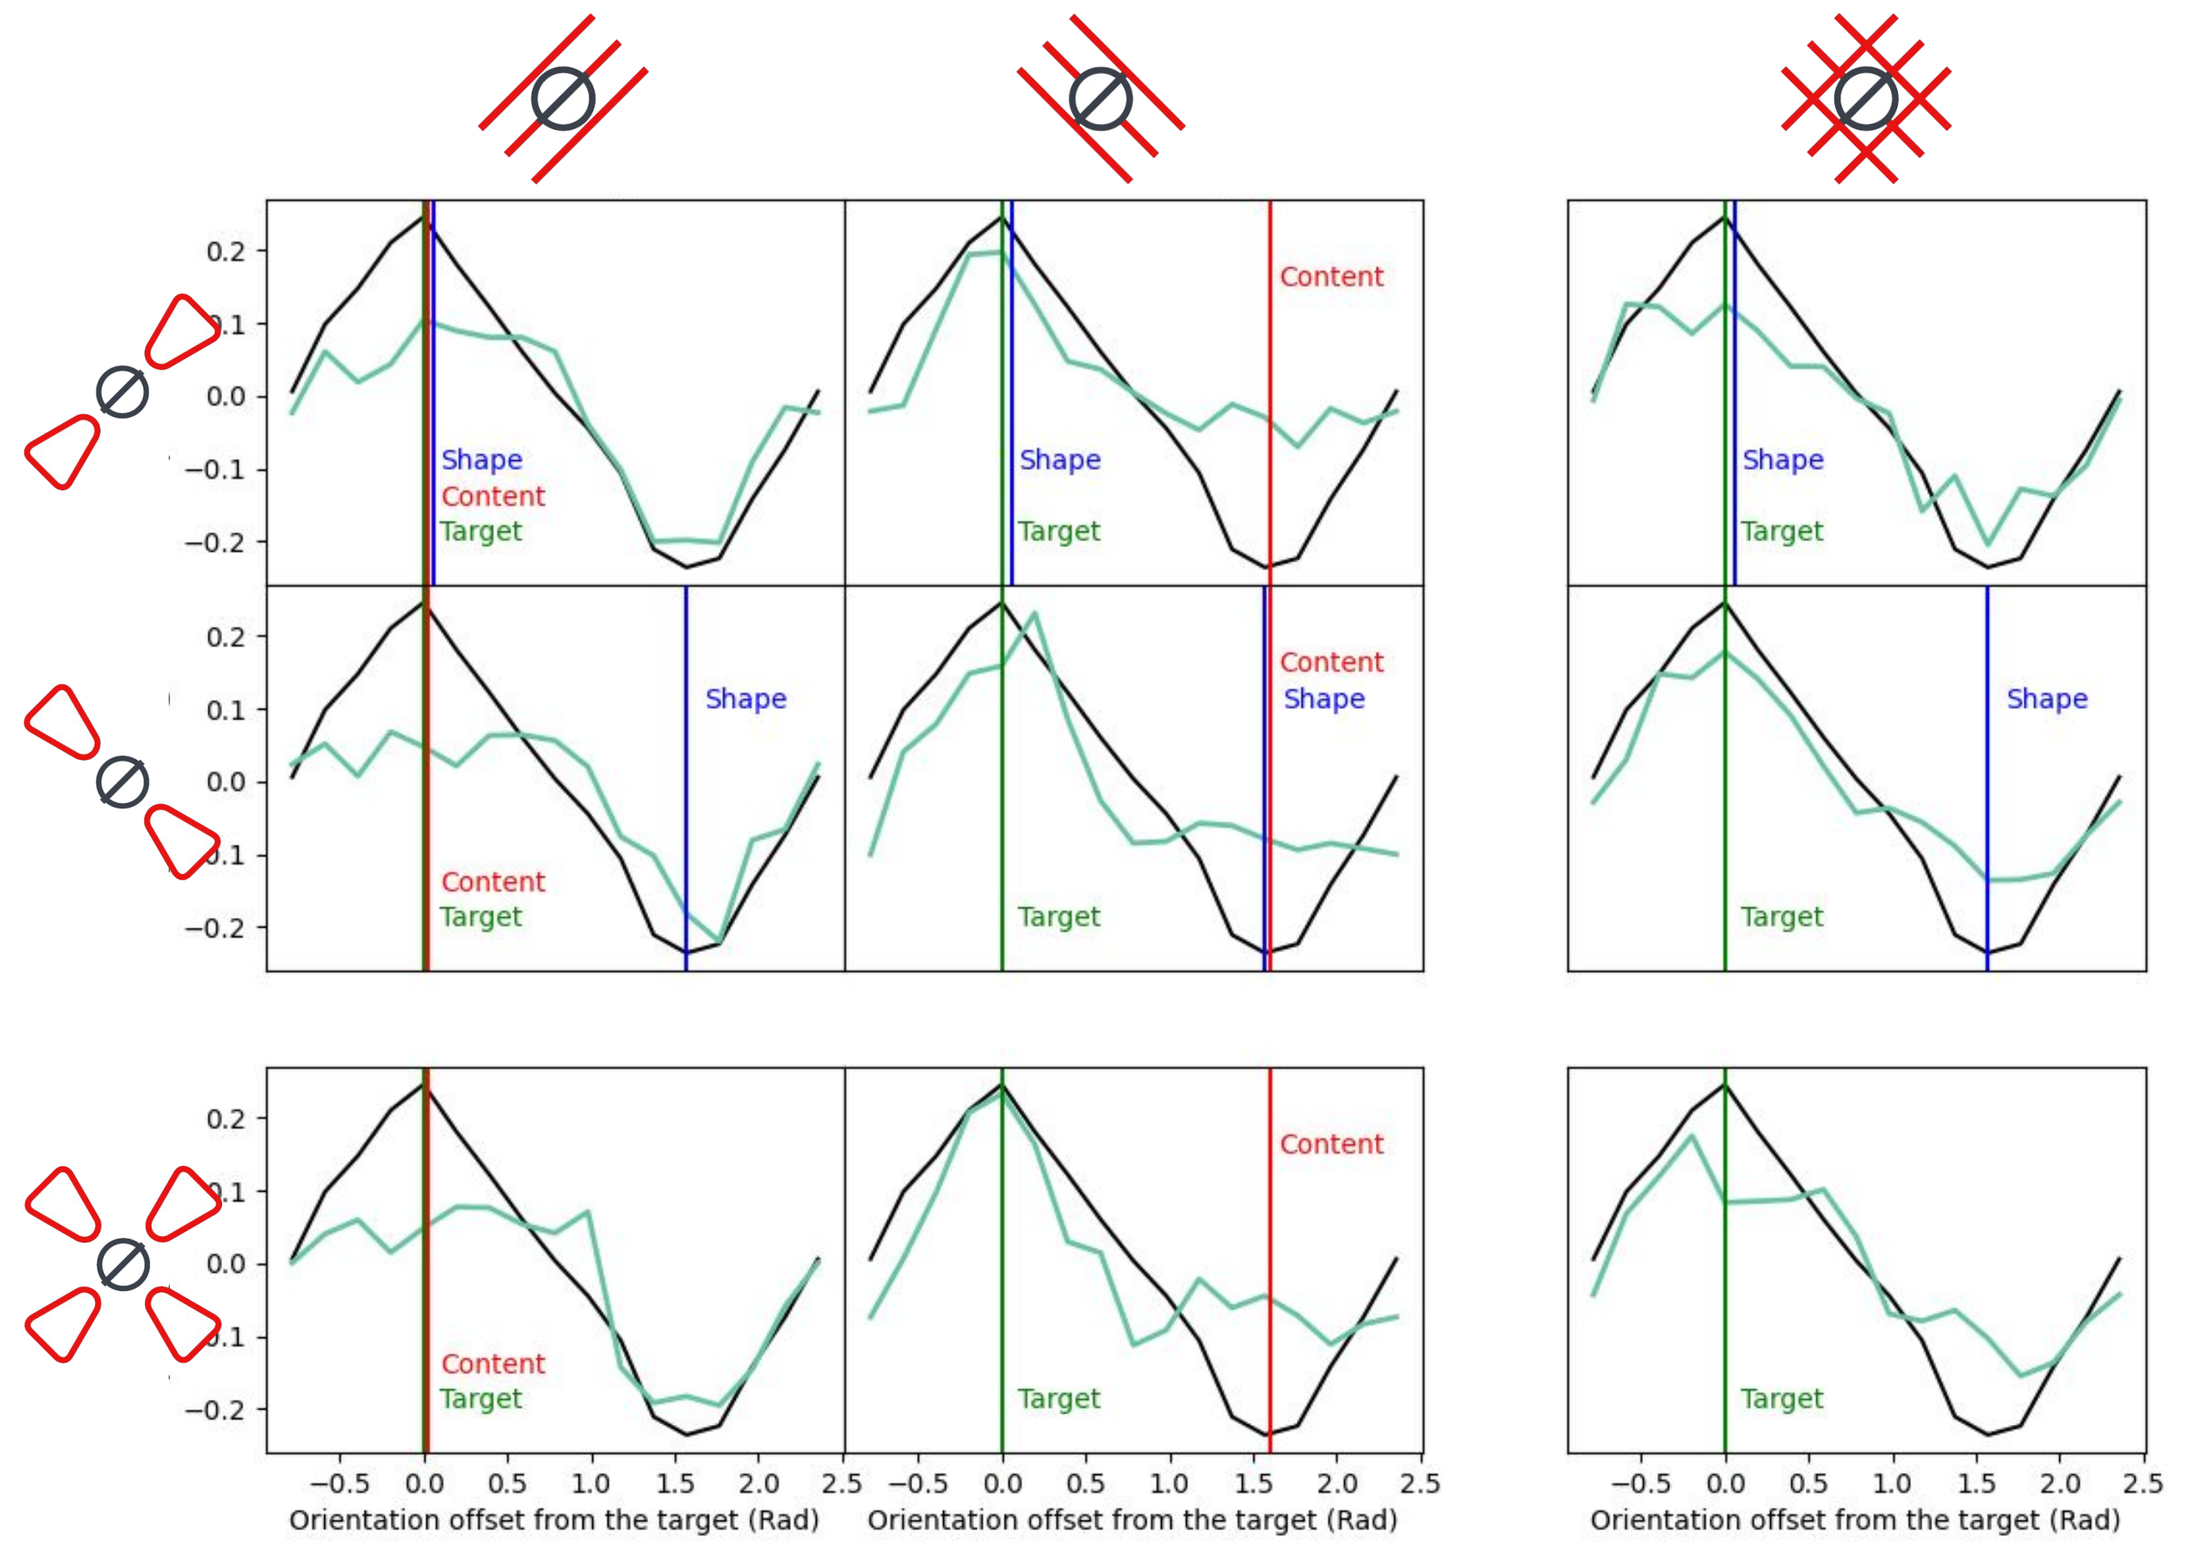

Shape
Content
Target
Orientation offset from the target (Rad)

So what?

Grating orientation has a linear, additive effect. **Global structure** of the surround only plays a smaller role.

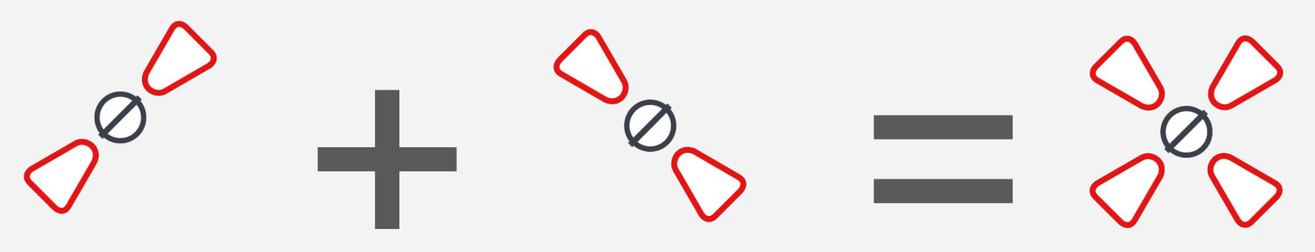

Performance

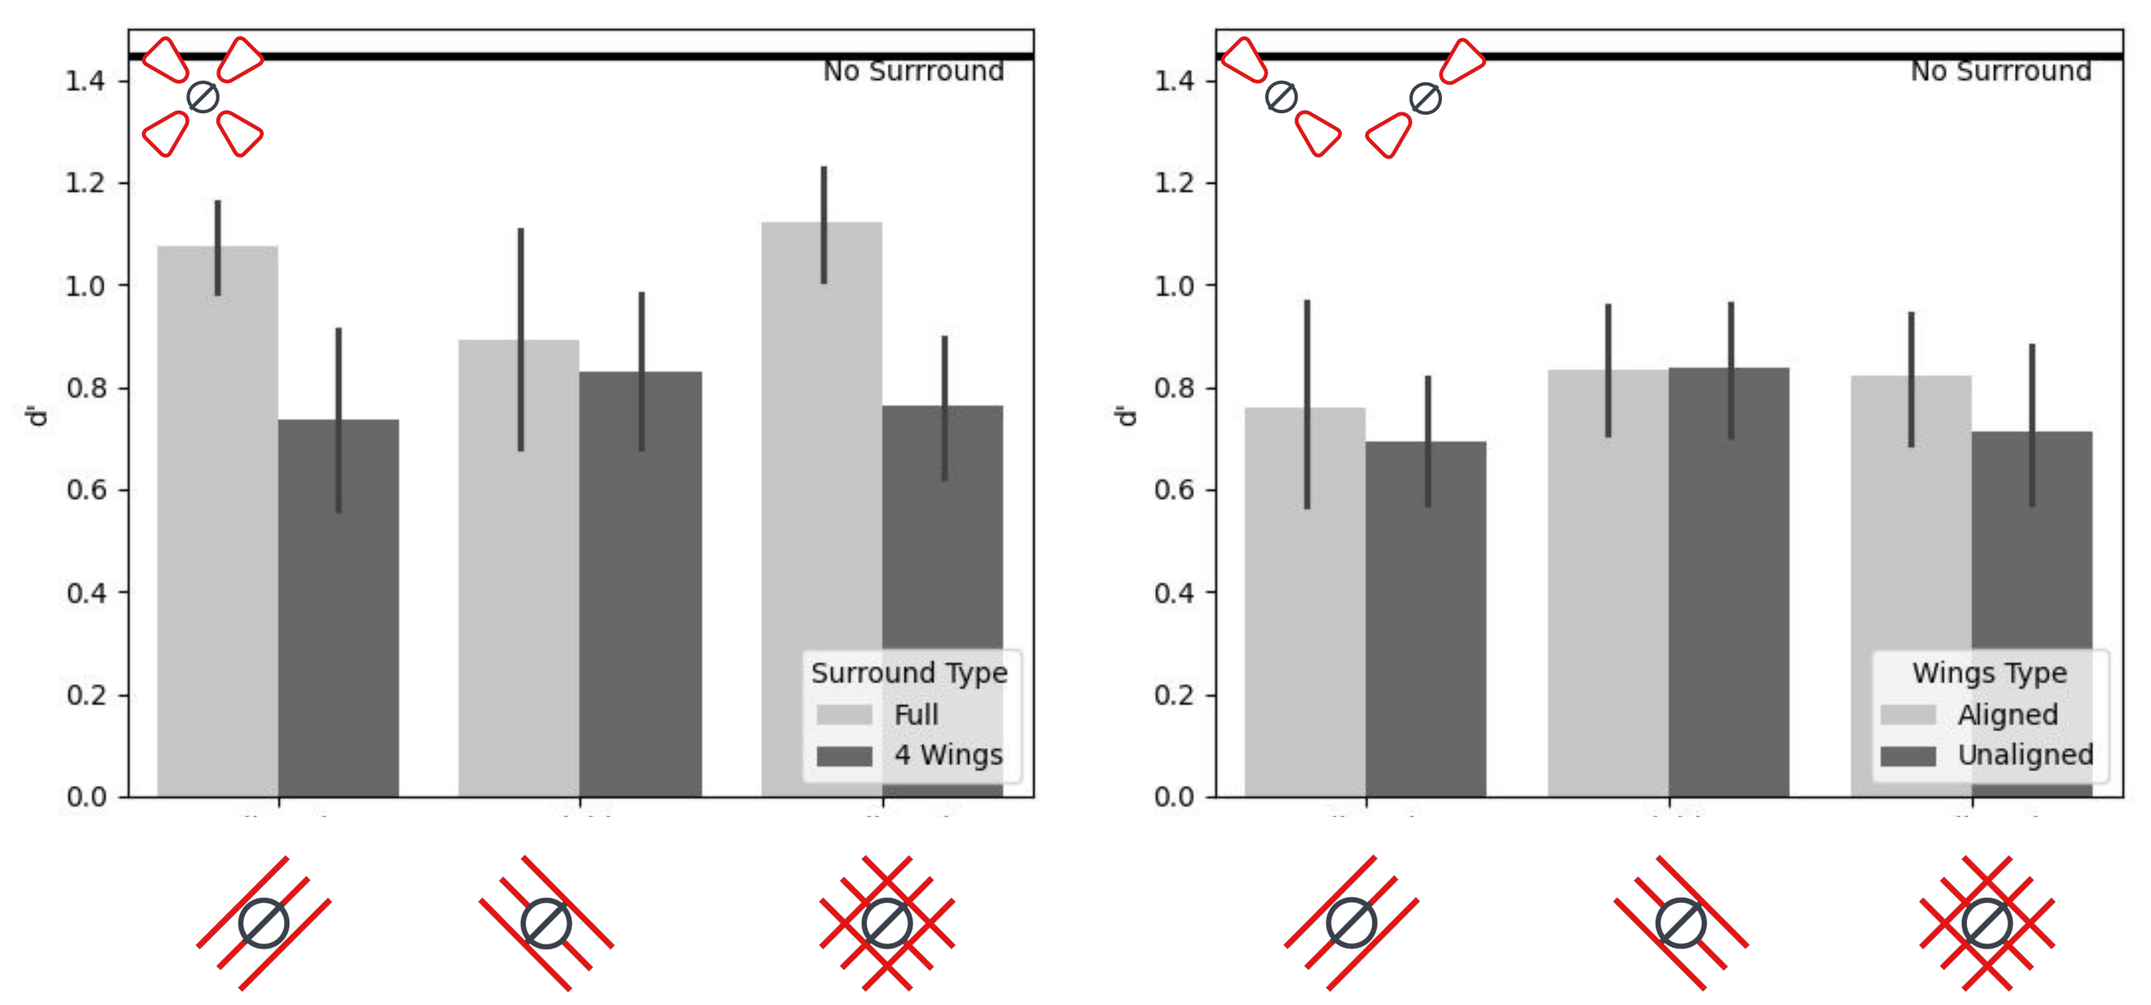

No Surround
d'
Surround Type
Full
4 Wings
Wings Type
Aligned
Unaligned

Temporal Context

Stimuli

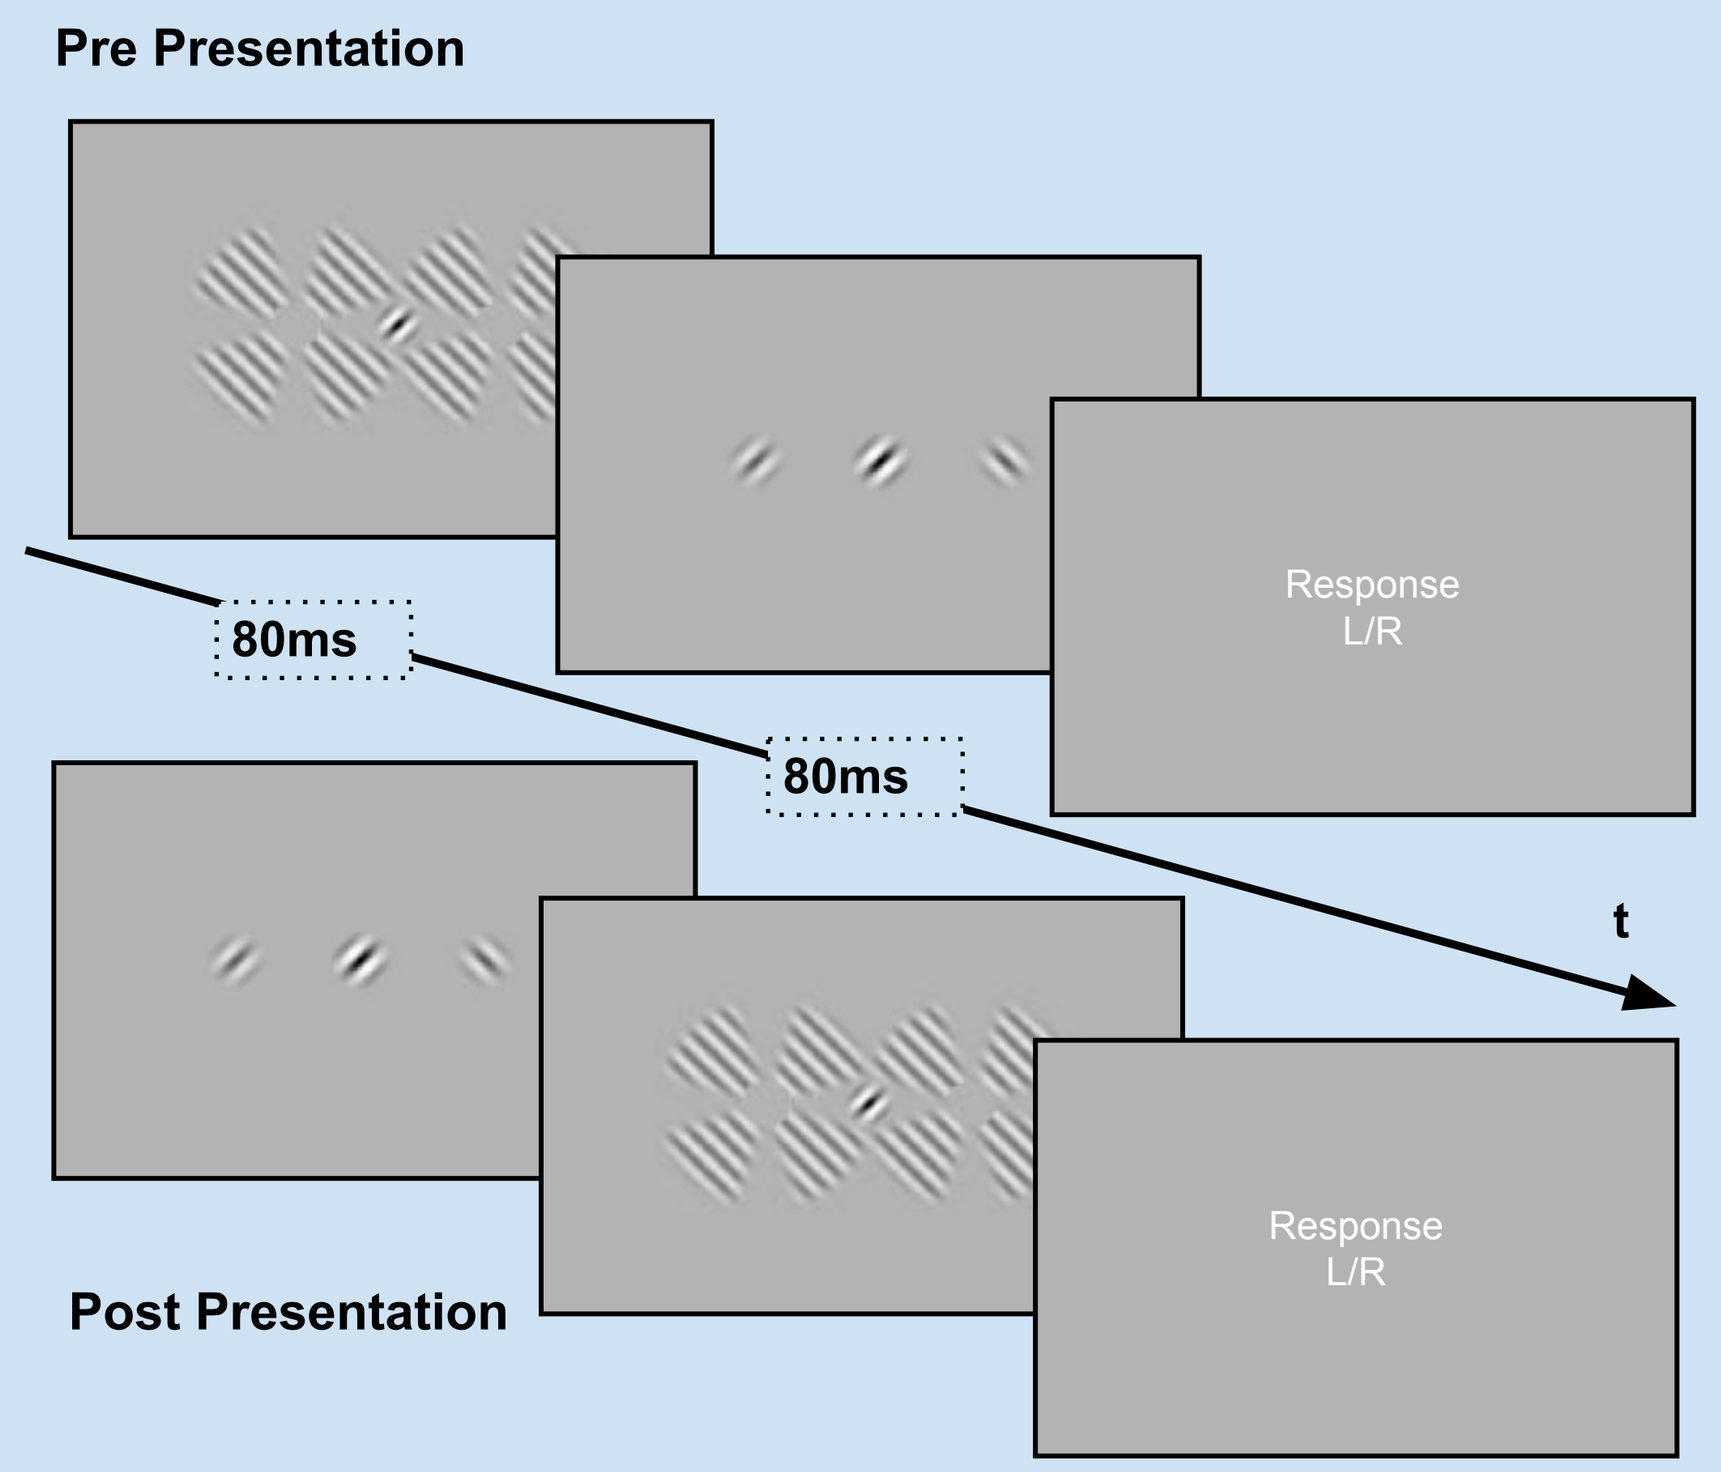

Pre Presentation
Response L/R
80ms
80ms
t
Post Presentation

Results

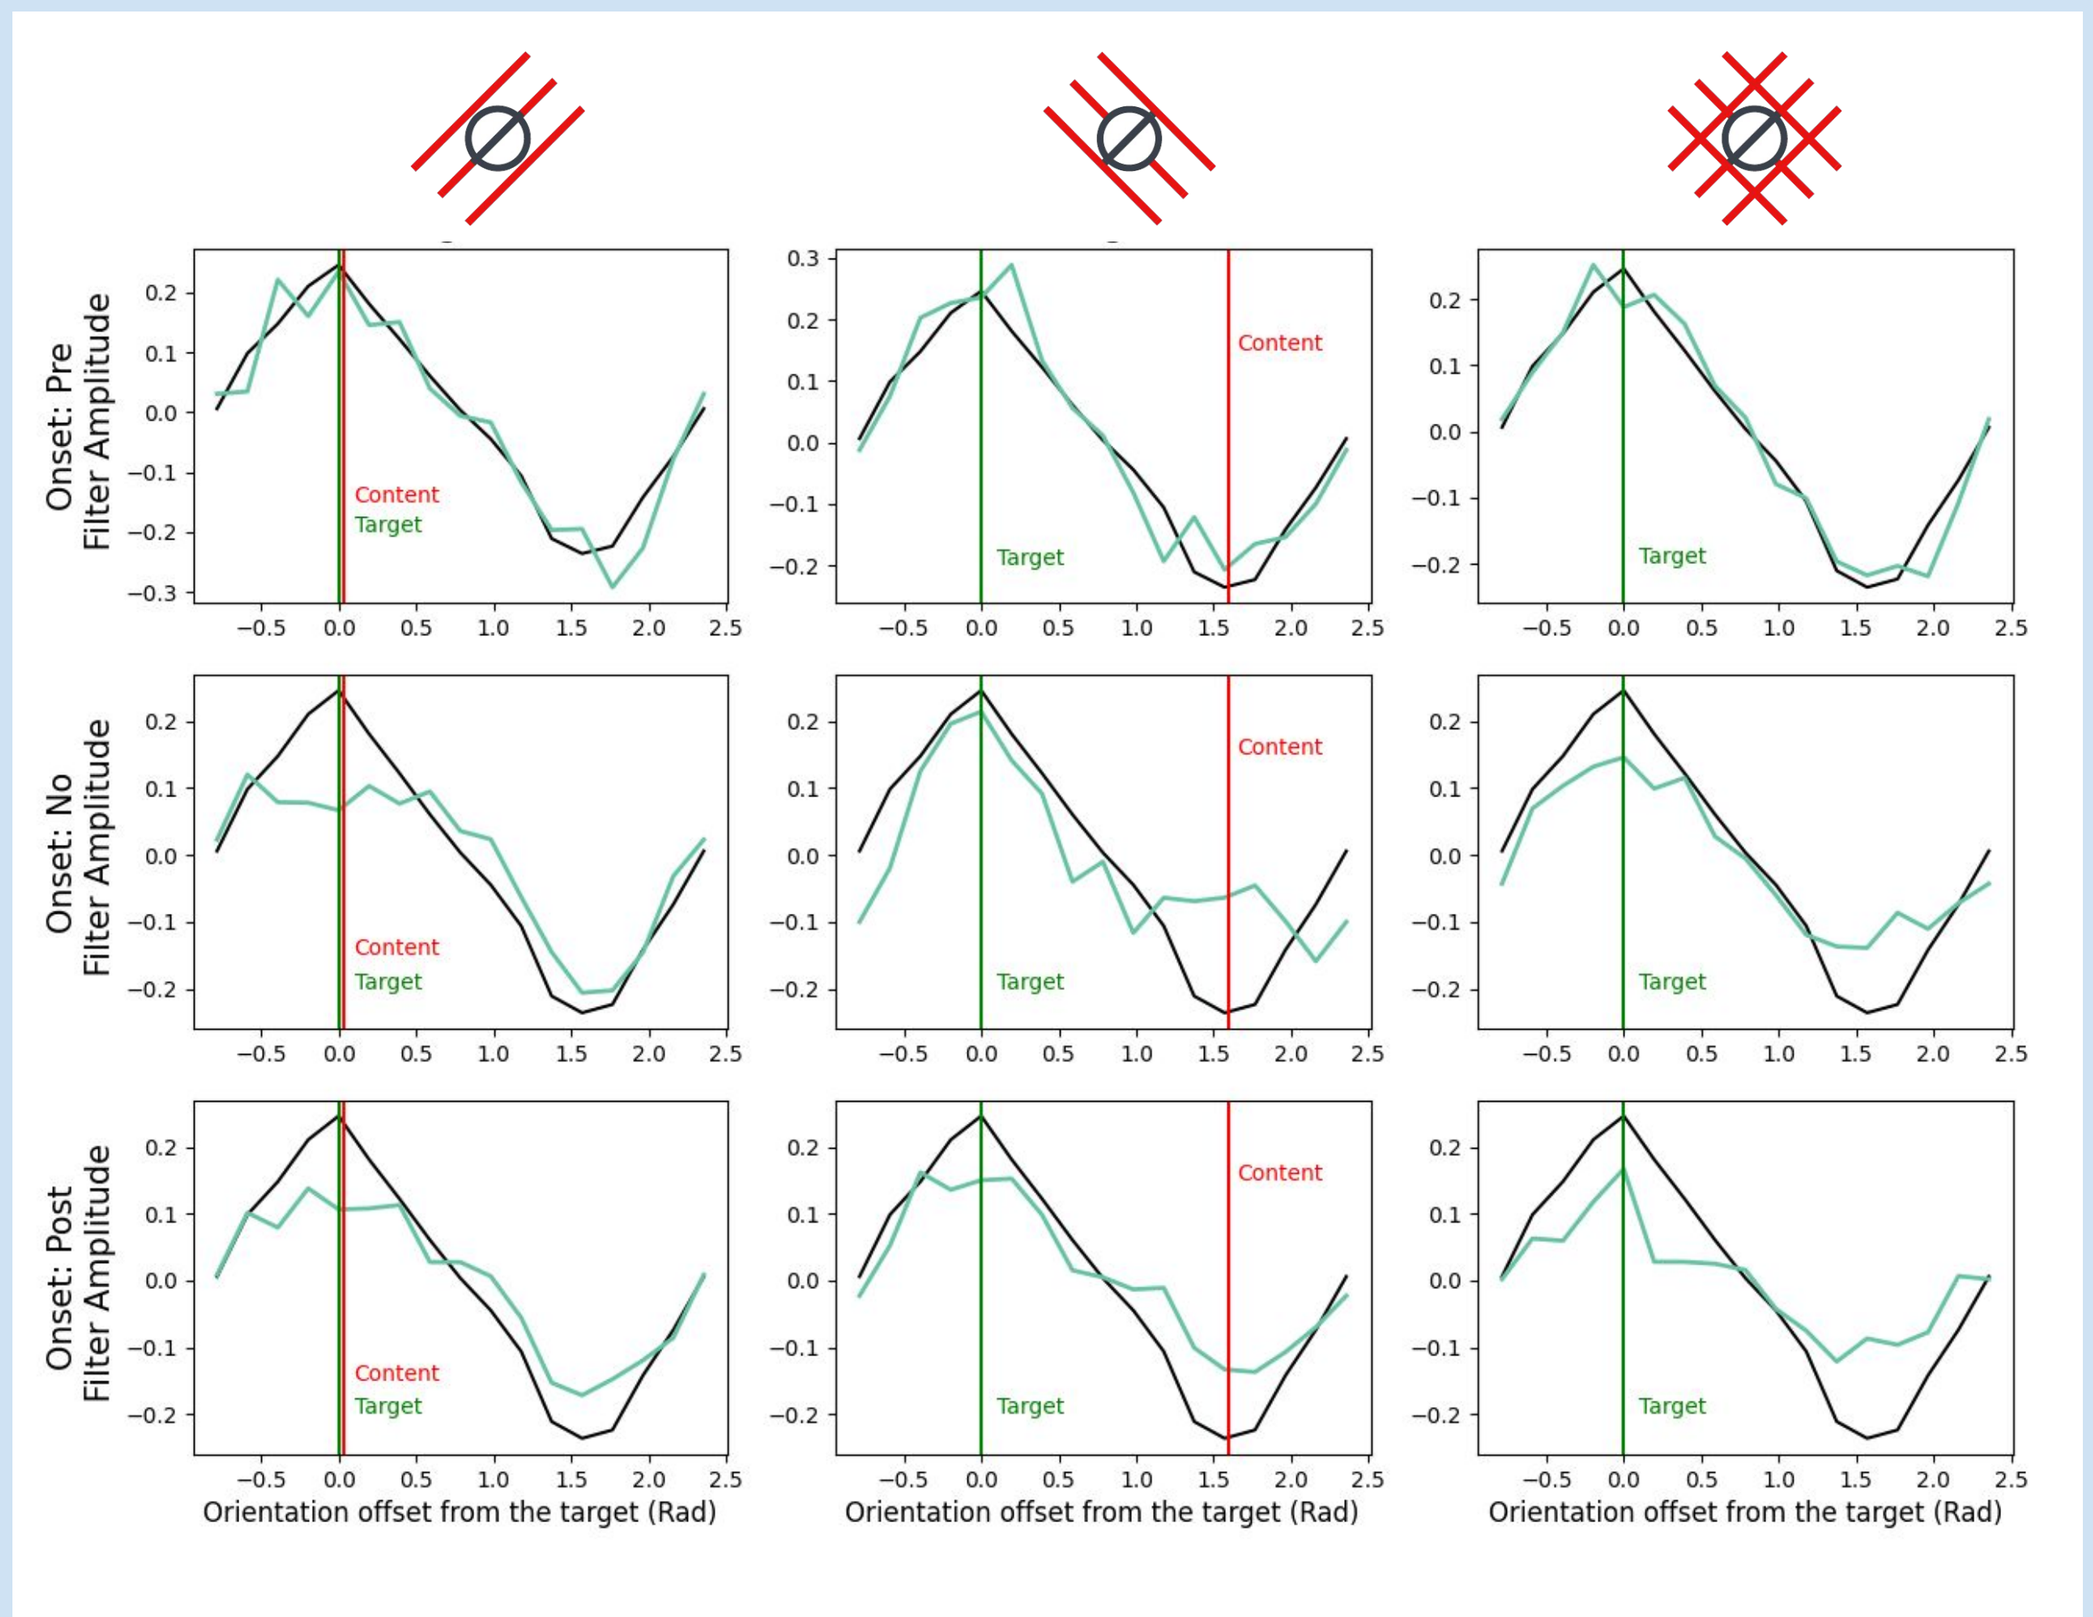

Onset: Pre Filter Amplitude
Onset: No Filter Amplitude
Onset: Post Filter Amplitude
Content
Target
Orientation offset from the target (Rad)

So what?

The effect of **Temporal context** is less linear. **Previews** and **postviews** affect orientation discrimination differently.

Performance

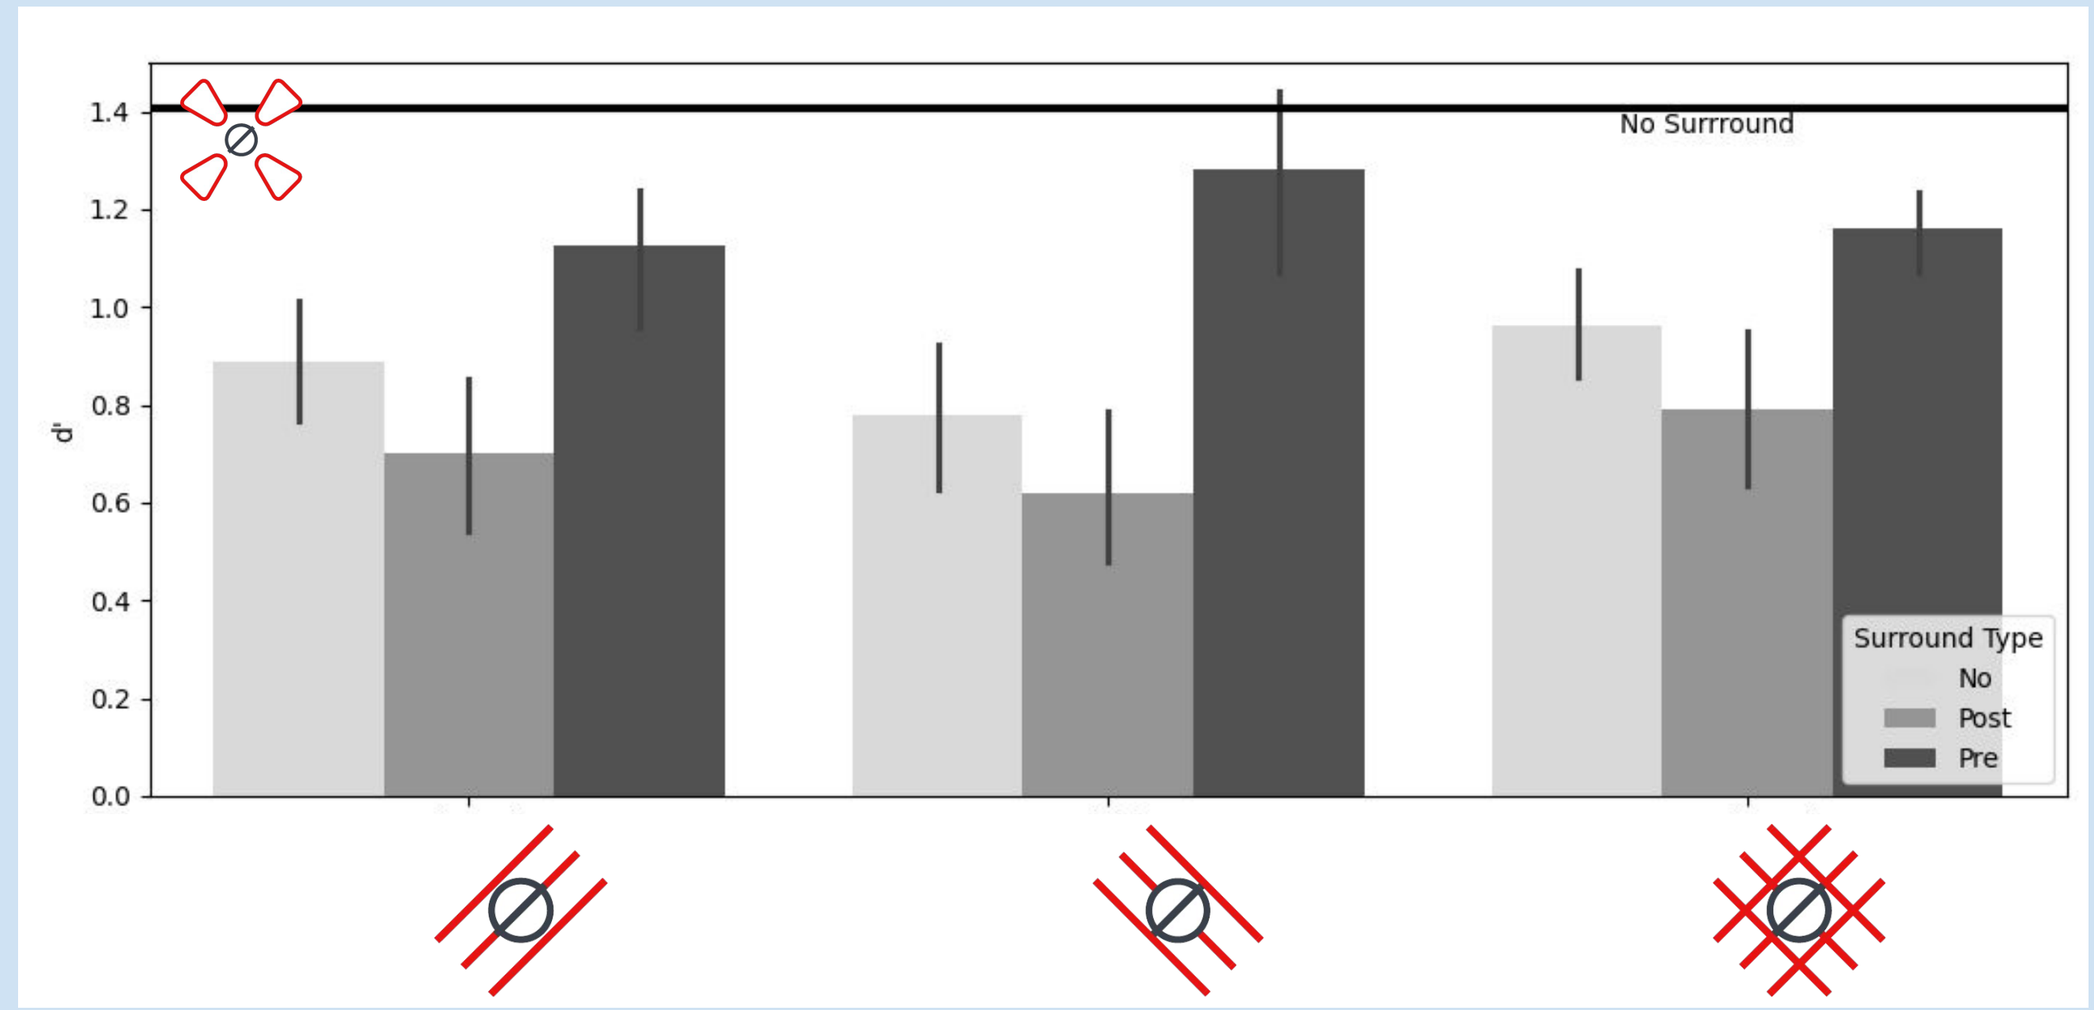

No Surround
d'
Surround Type
No
Post
Pre