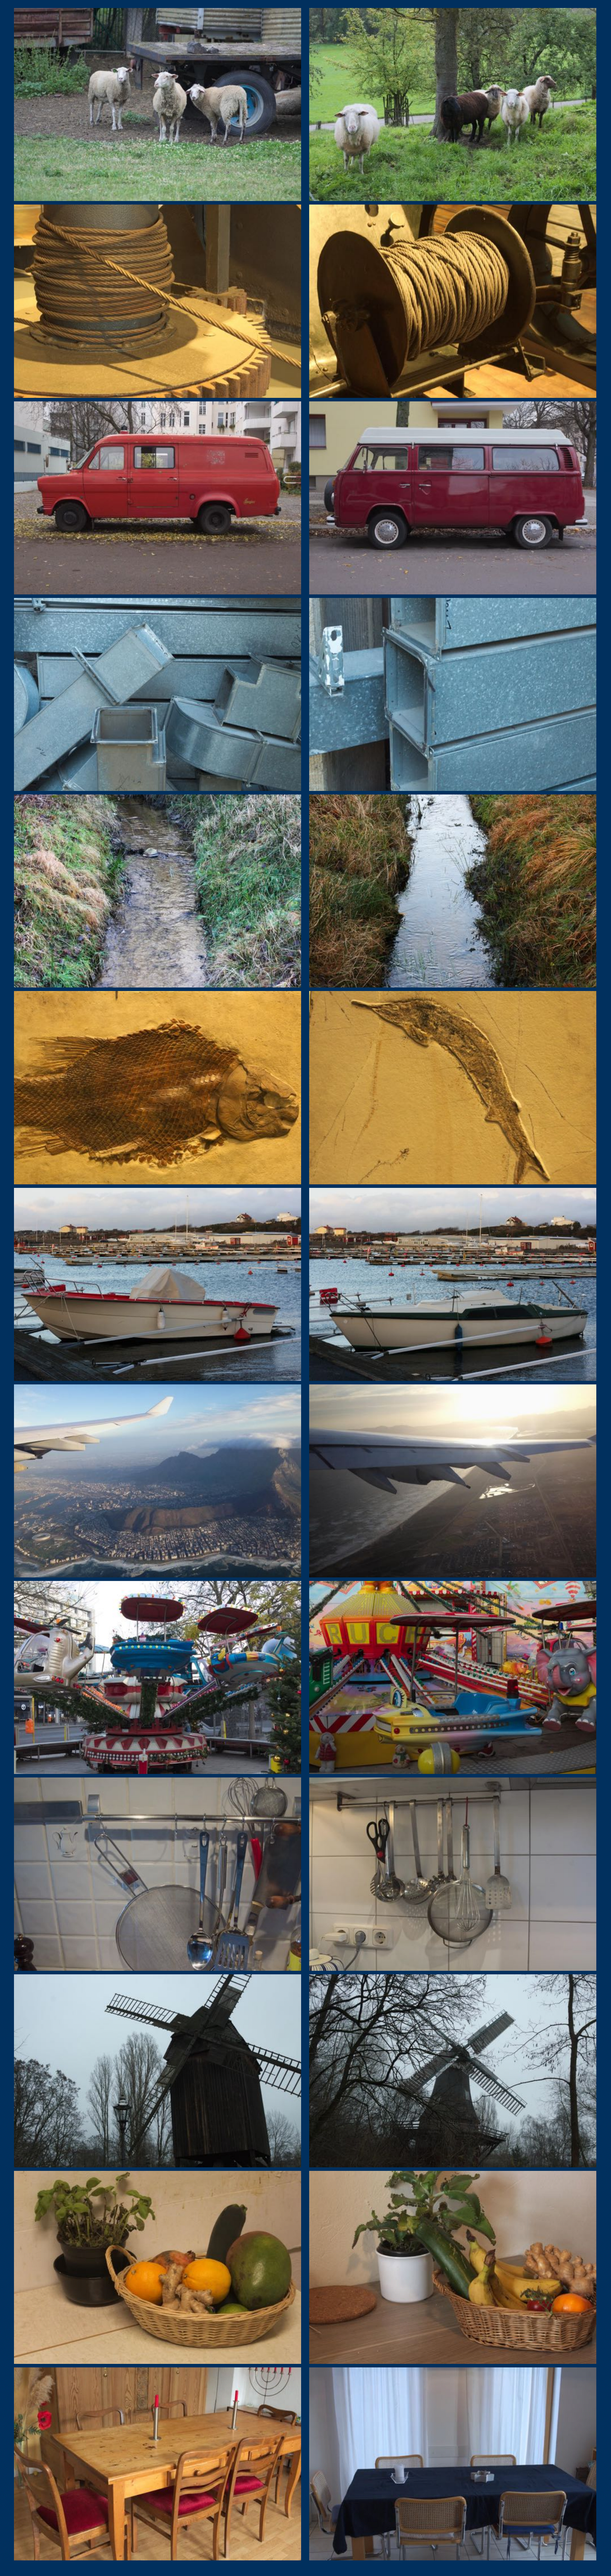

The Effect of Visual Long-Term Memory on Eye Movements over Time

Lisa F. Schwetlick, Hans A. Trukenbrod & Ralf Engbert

Introduction

- Familiarity with an image can affect eye movement measures:
 - in visual search[1]: speed and performance benefit
 - in free viewing[2,3,4]: **fixation duration increases** and **saccade amplitude decreases**
- Capacity and persistence of visual long-term memory (VLTM) [5] is robust: images stay "familiar" over long periods
- The effects of VLTM on eye movement have only been investigated on **short time scales** (seconds or minutes; repeated presentation within the same session)
- Do these image familiarity effects on eye movement **transfer to longer time scales**?

Method

- We present a series of 3 experiments which varied the time between repetitions:
 - Experiment 1: 32 participants viewed images in 3 sessions with several days between sessions
 - Experiment 2: 30 participants viewed images in 3 sessions with session 1 and 2 on the same day and session 3 on the next day
 - Experiment 3: 32 participants viewed images in 1 session
- Data were recorded with an Eyelink 1000 Eyetracker (SR Research)
- Images could be unfamiliar, familiar or semantically familiar
- We generated a new image set of 188 pairs of similar images
- Session 3 in Experiment 1 and 2 employed a gaze contingent viewing paradigm (periphery appears scrambled) to investigate memory-driven behavior in the absence of peripheral information

Experiment 1 / Experiment 2

Session 1: *View (8s):* 106 new images. *Recall:* 8 familiar images, 8 new images, 8 semantically related images

days (Experiment 1) / hours (Experiment 2)

Session 2: *View (8s):* 46 new images, 30 familiar images, 30 semantically related images. *Recall:* 8 familiar, 8 new, 8 semantically related images

days (Experiment 1) / hours (Experiment 2)

Session 3: *Gaze Contingent viewing (10s) and respond:* 20 new images, 20 seen twice, 20 seen once, 20 images seen once and similar

Experiment 3

Training: *View (8s):* 10 new images. *Recall:* 5 familiar images, 5 new images, 5 semantically related images

minutes

Main: *View (8s):* 60 new images, 30 familiar images, 30 semantically related images. *Recall:* 5 seen twice, 5 seen and seen sim, 5 seen sim, 5 new

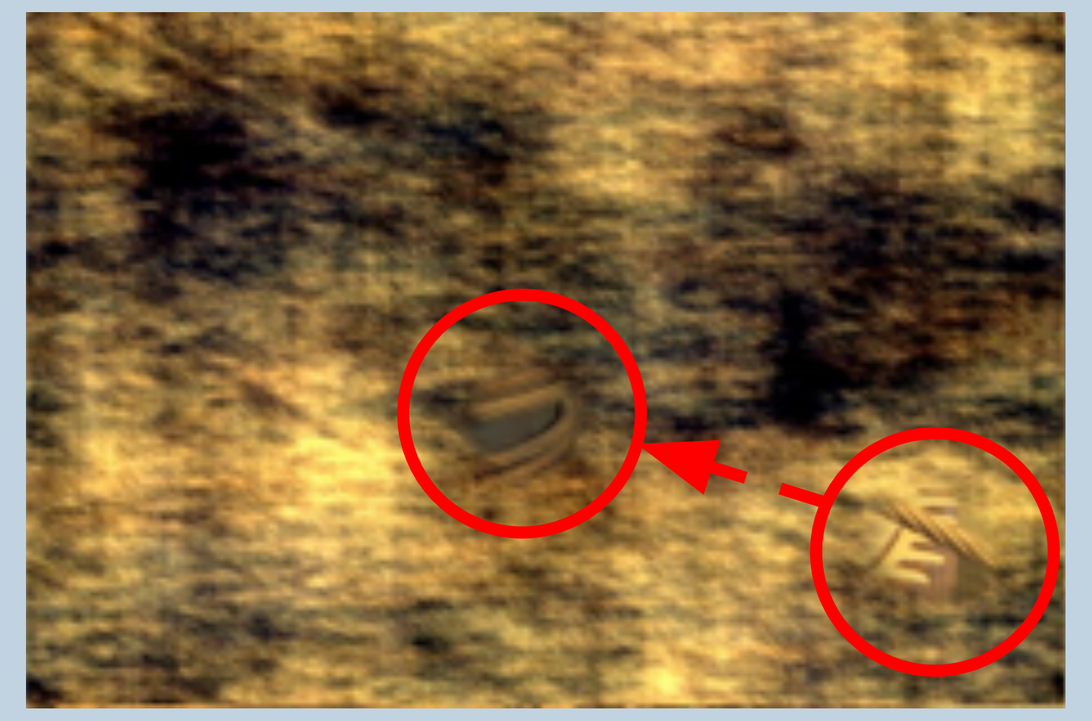

Results: Regular Viewing

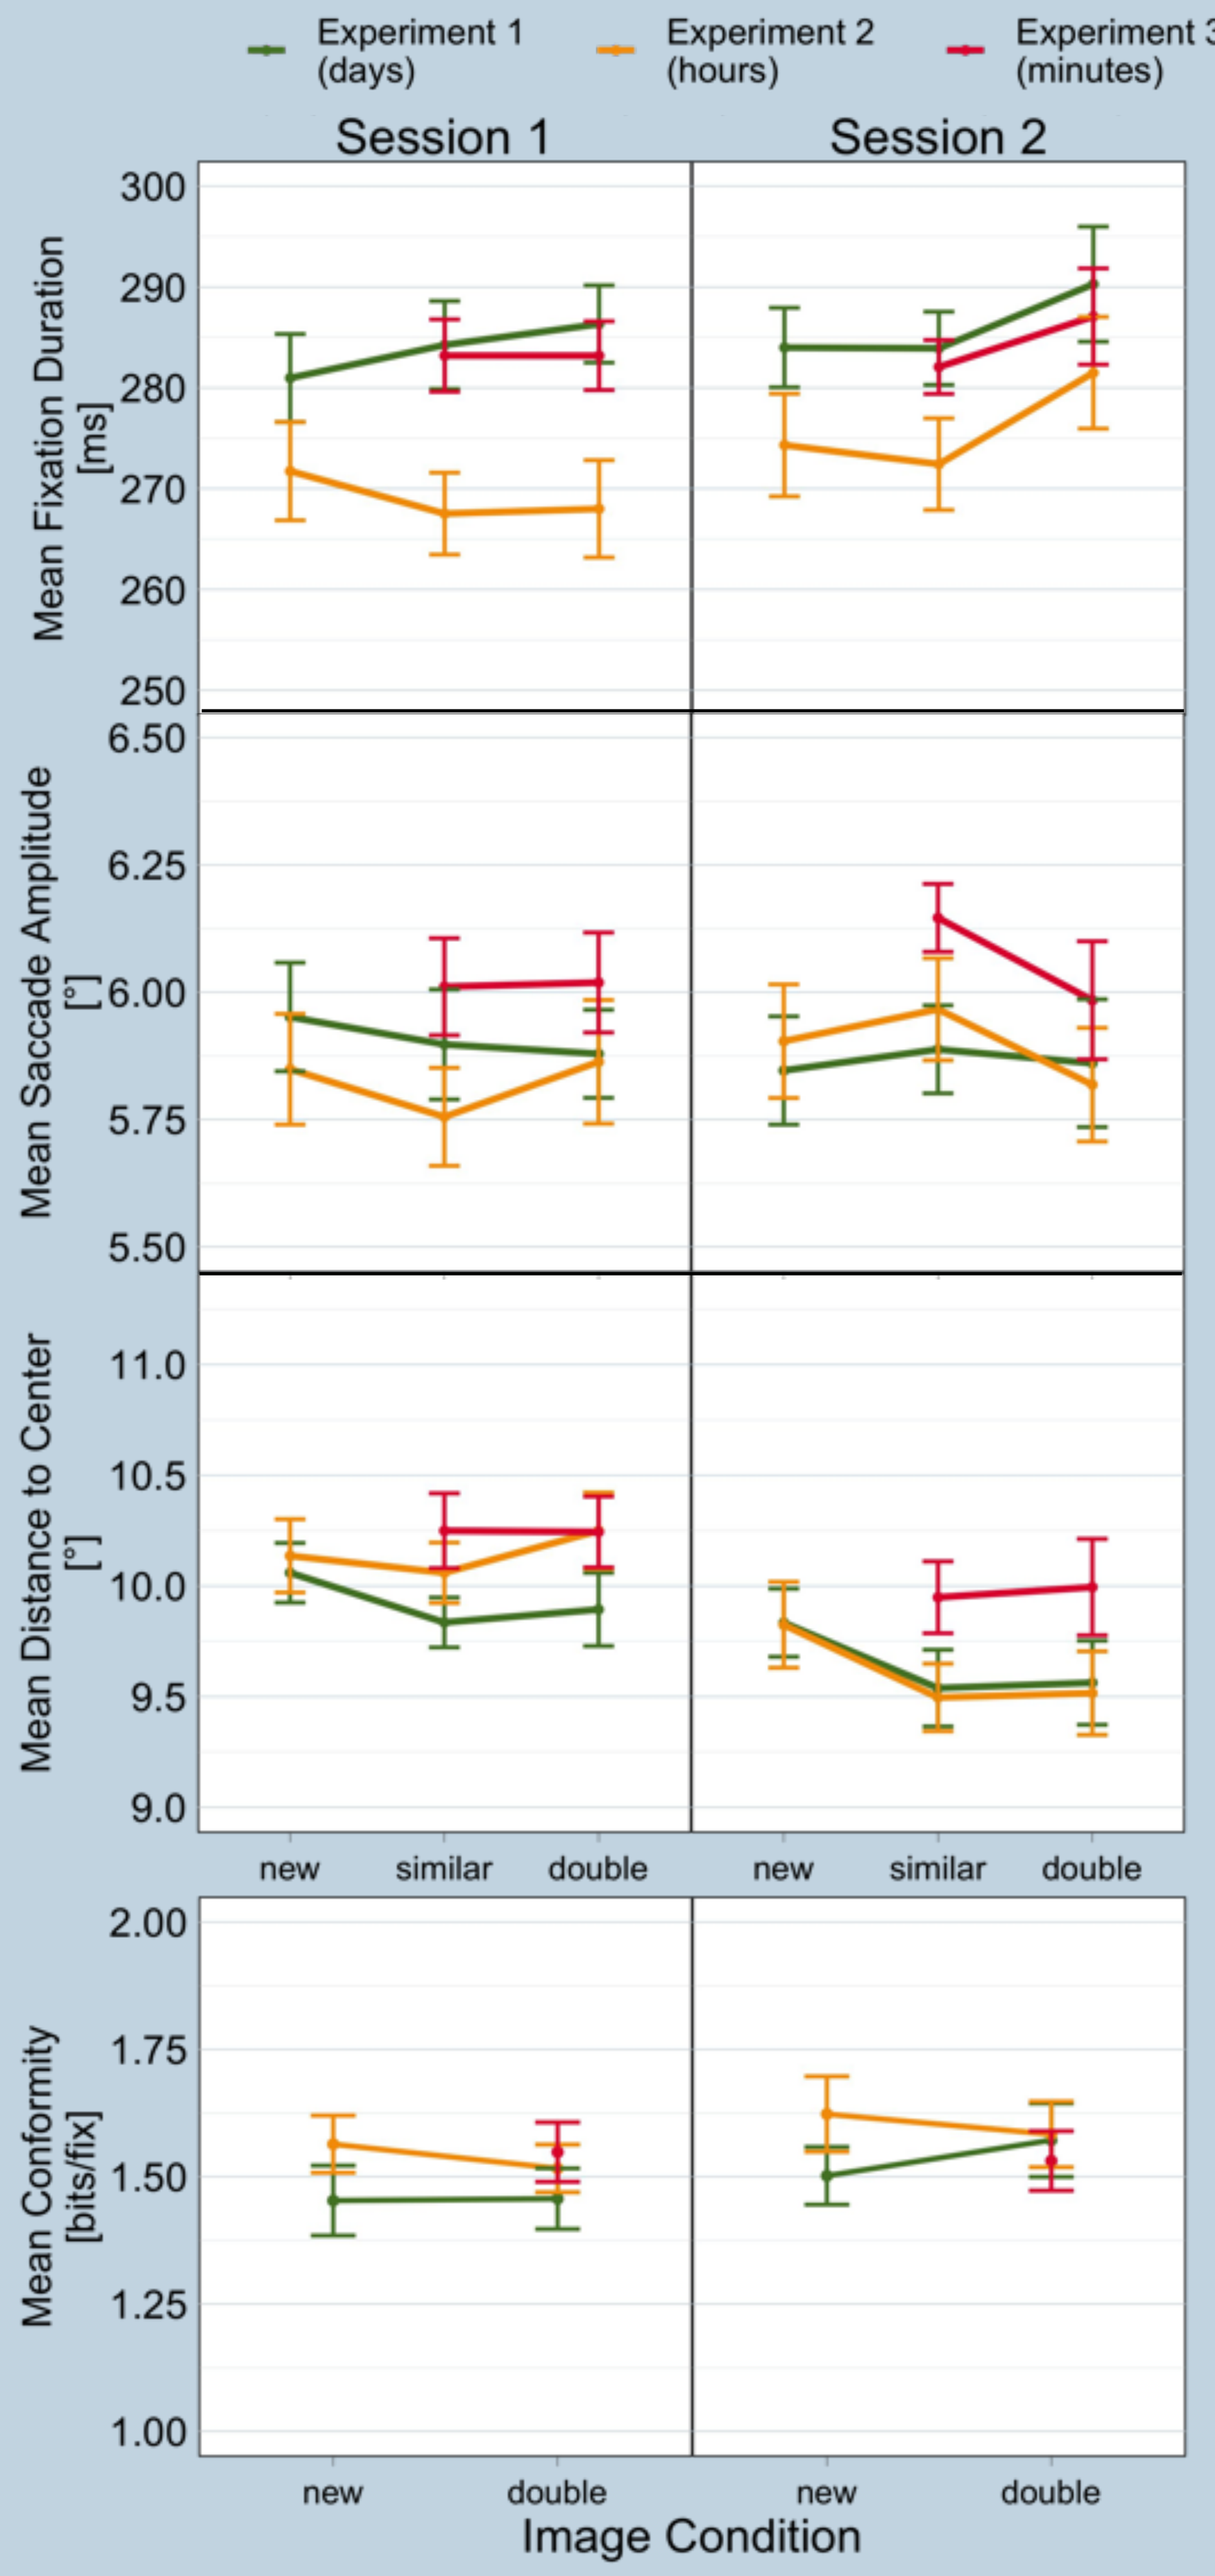

Experiment 1 (days)
Experiment 2 (hours)
Experiment 3 (minutes)
Session 1
Session 2
Mean Fixation Duration [ms]
Mean Saccade Amplitude [°]
Mean Distance to Center [°]
Mean Conformity [bits/fix]
new
similar
double
Image Condition

Fixation Duration
Statistically we were not able to replicate the prolongation of fixation durations for the second viewing.

Saccade Amplitude
Statistically we were not able to replicate the reduction of saccade amplitudes for the second viewing.

Distance to Center
Observers showed a stronger tendency to fixate the center during the second viewing of an image.

Conformity/ Fixation Likelihood
Observers did not show increses in scanpath predictability during the second viewing of an image.

Results: Gaze Contingent Viewing

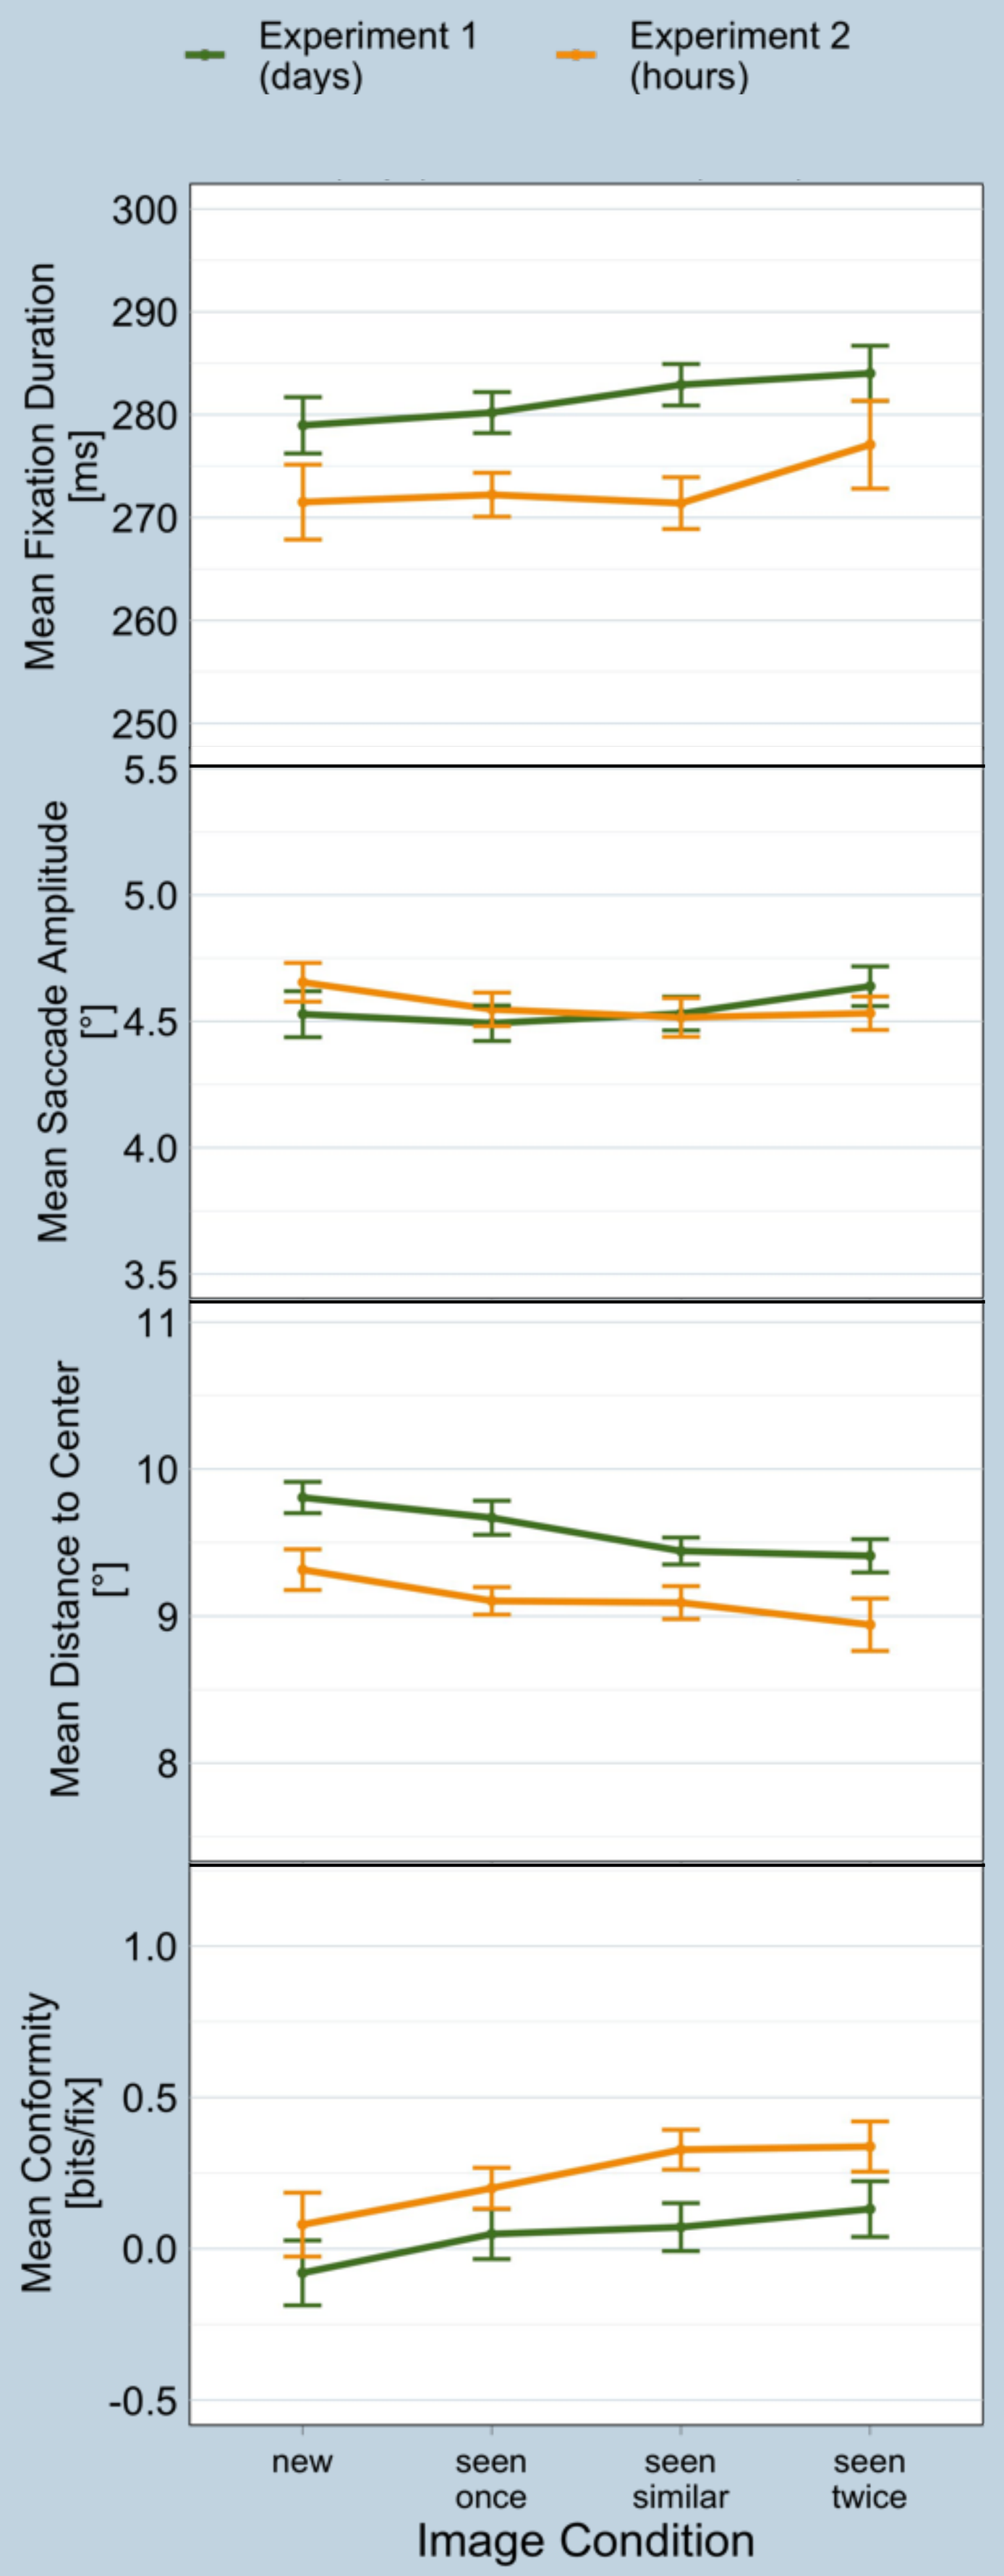

Experiment 1 (days)
Experiment 2 (hours)
Mean Fixation Duration [ms]
Mean Saccade Amplitude [°]
Mean Distance to Center [°]
Mean Conformity [bits/fix]
new
seen once
seen similar
seen twice
Image Condition

Fixation Duration
In the absence of peripheral information fixation duration increases slightly, but not significantly

Saccade Amplitude:
We found no significant change in saccade amplitude depending on image familiarity.

Distance to Center
The tendency to fixate near the image center becomes less the more familiar the image.

Conformity/ Fixation Likelihood
The predictability of the scanpath increases marginally, when the image is familiar. Although overall predictability is low.

Results: Recognition

Session 1 and 2:

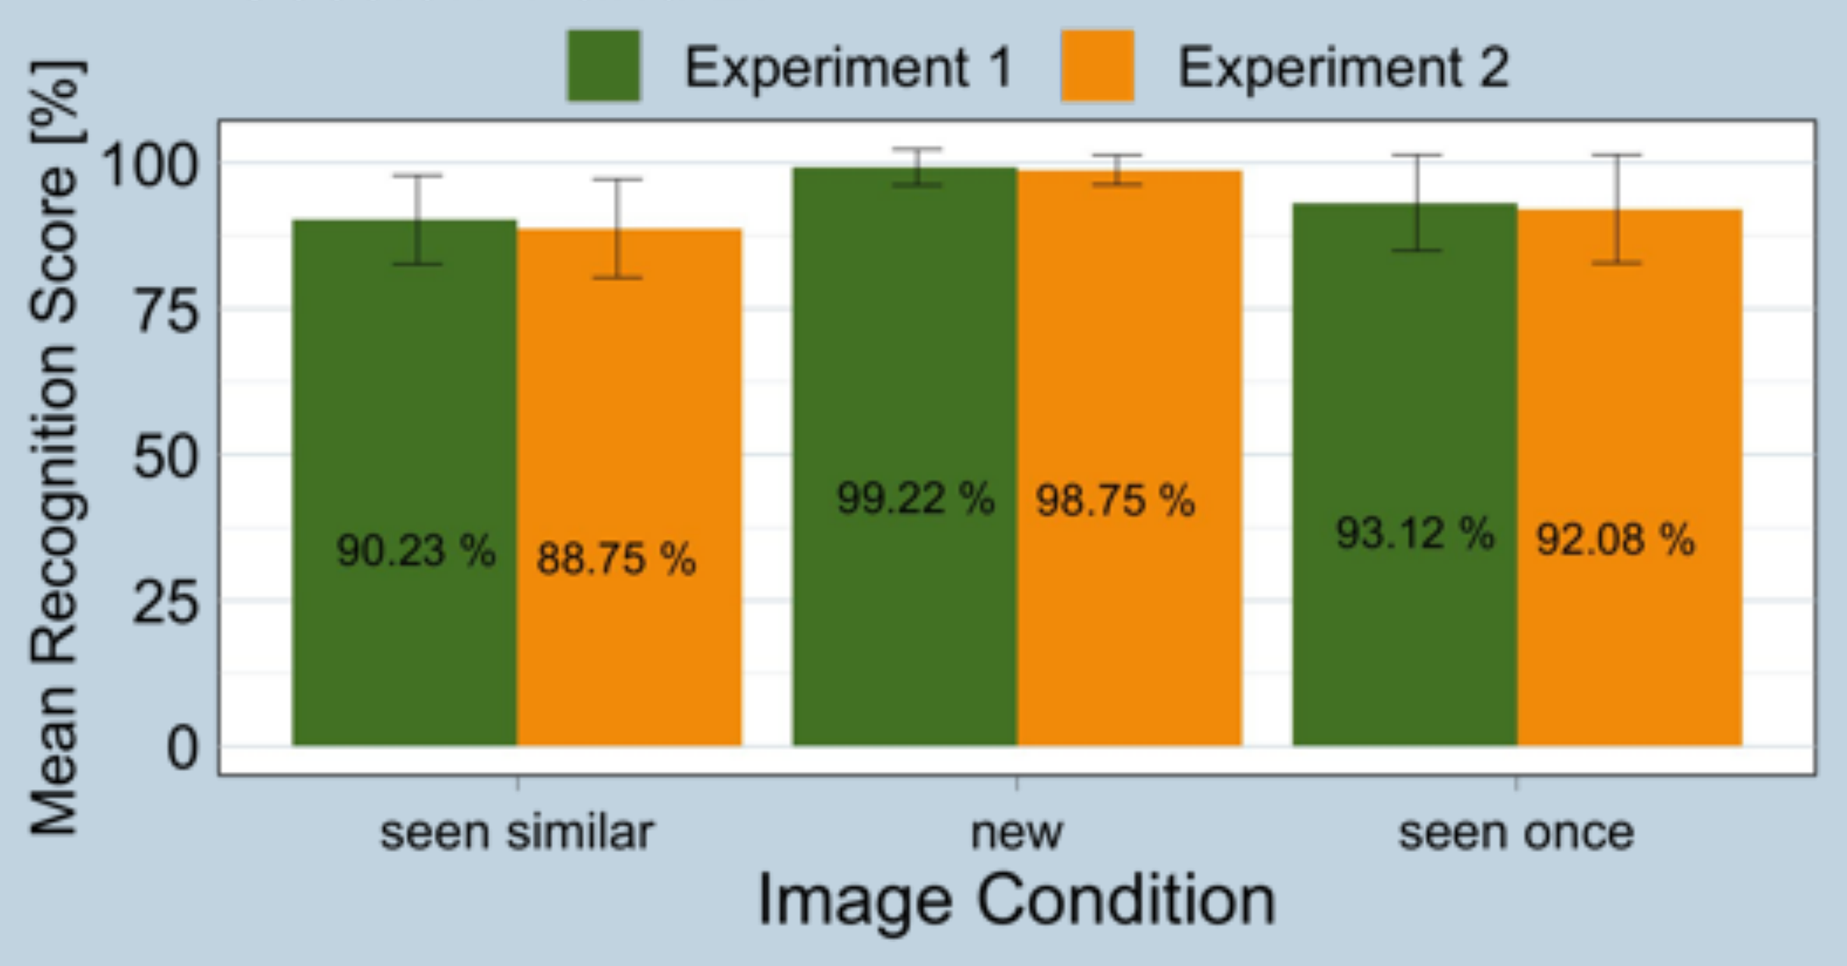

Experiment 1
Experiment 2
Mean Recognition Score [%]
90.23 %
88.75 %
99.22 %
98.75 %
93.12 %
92.08 %
seen similar
new
seen once
Image Condition

- Participants showed the expected proficiency in recognizing images
- The absence or reduction of the expected effects is therefore not due to a participant's failure to recognize the images.

Session 3:

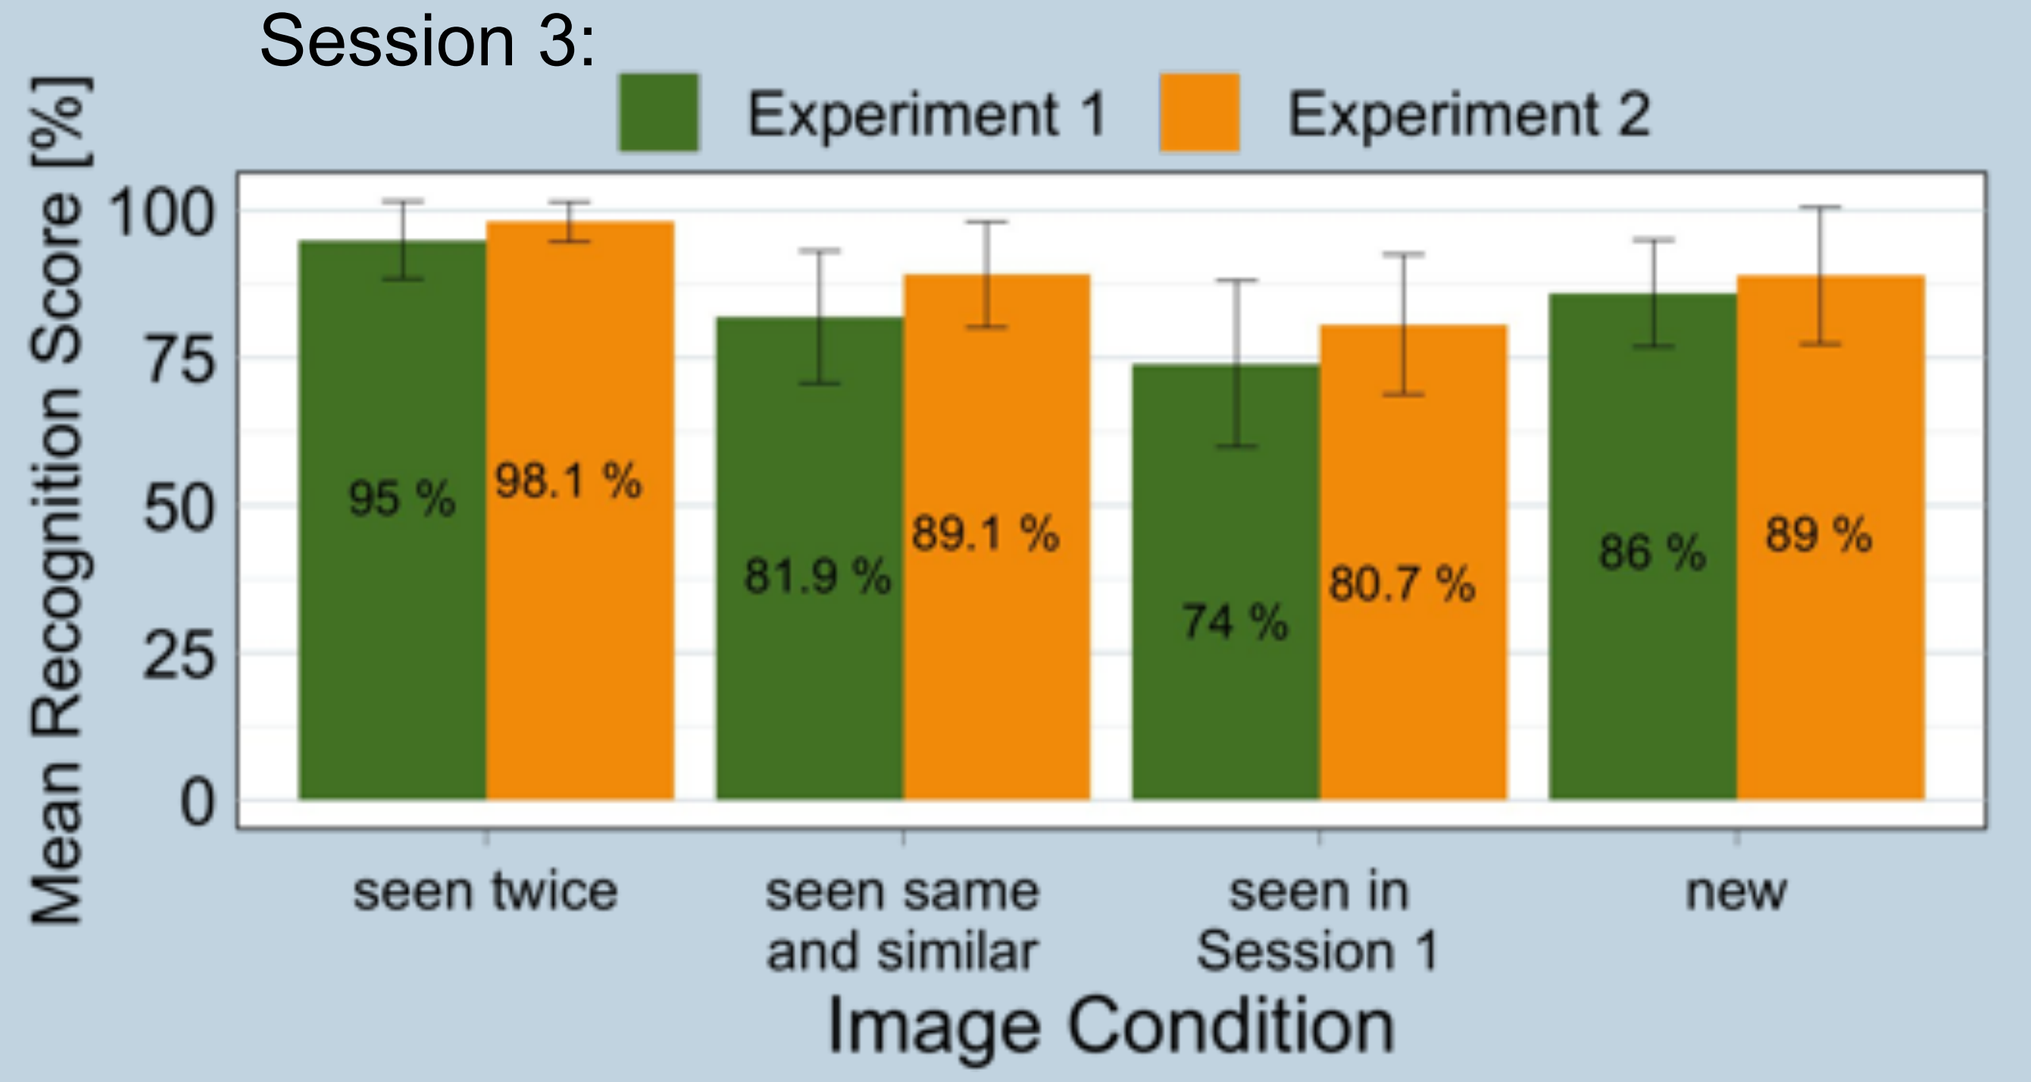

Experiment 1
Experiment 2
Mean Recognition Score [%]
95 %
98.1 %
81.9 %
89.1 %
74 %
80.7 %
86 %
89 %
seen twice
seen same and similar
seen in Session 1
new
Image Condition

Conformity: Fixation Likelihood

$$conformity = \frac{1}{N}\sum_{i=1}^{N} \log_2(d_j(x_i))$$

$x =$ Fixation Position

$d_j =$ density of image j

$i - N =$ Fixation number

Viewed image *j*

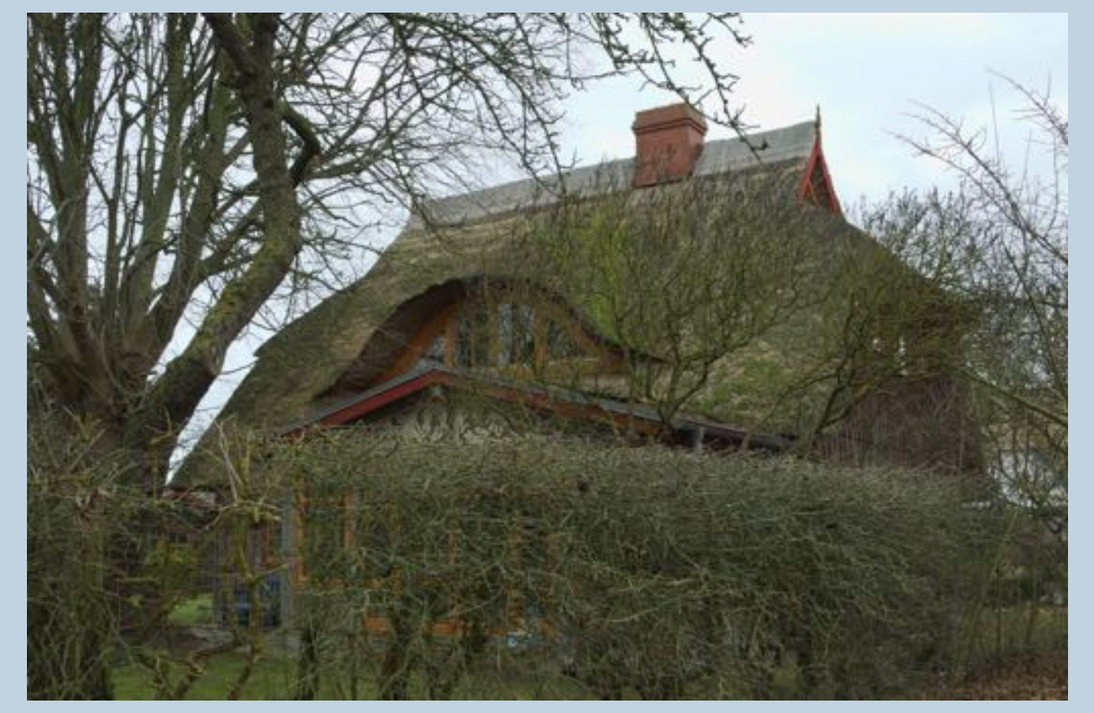

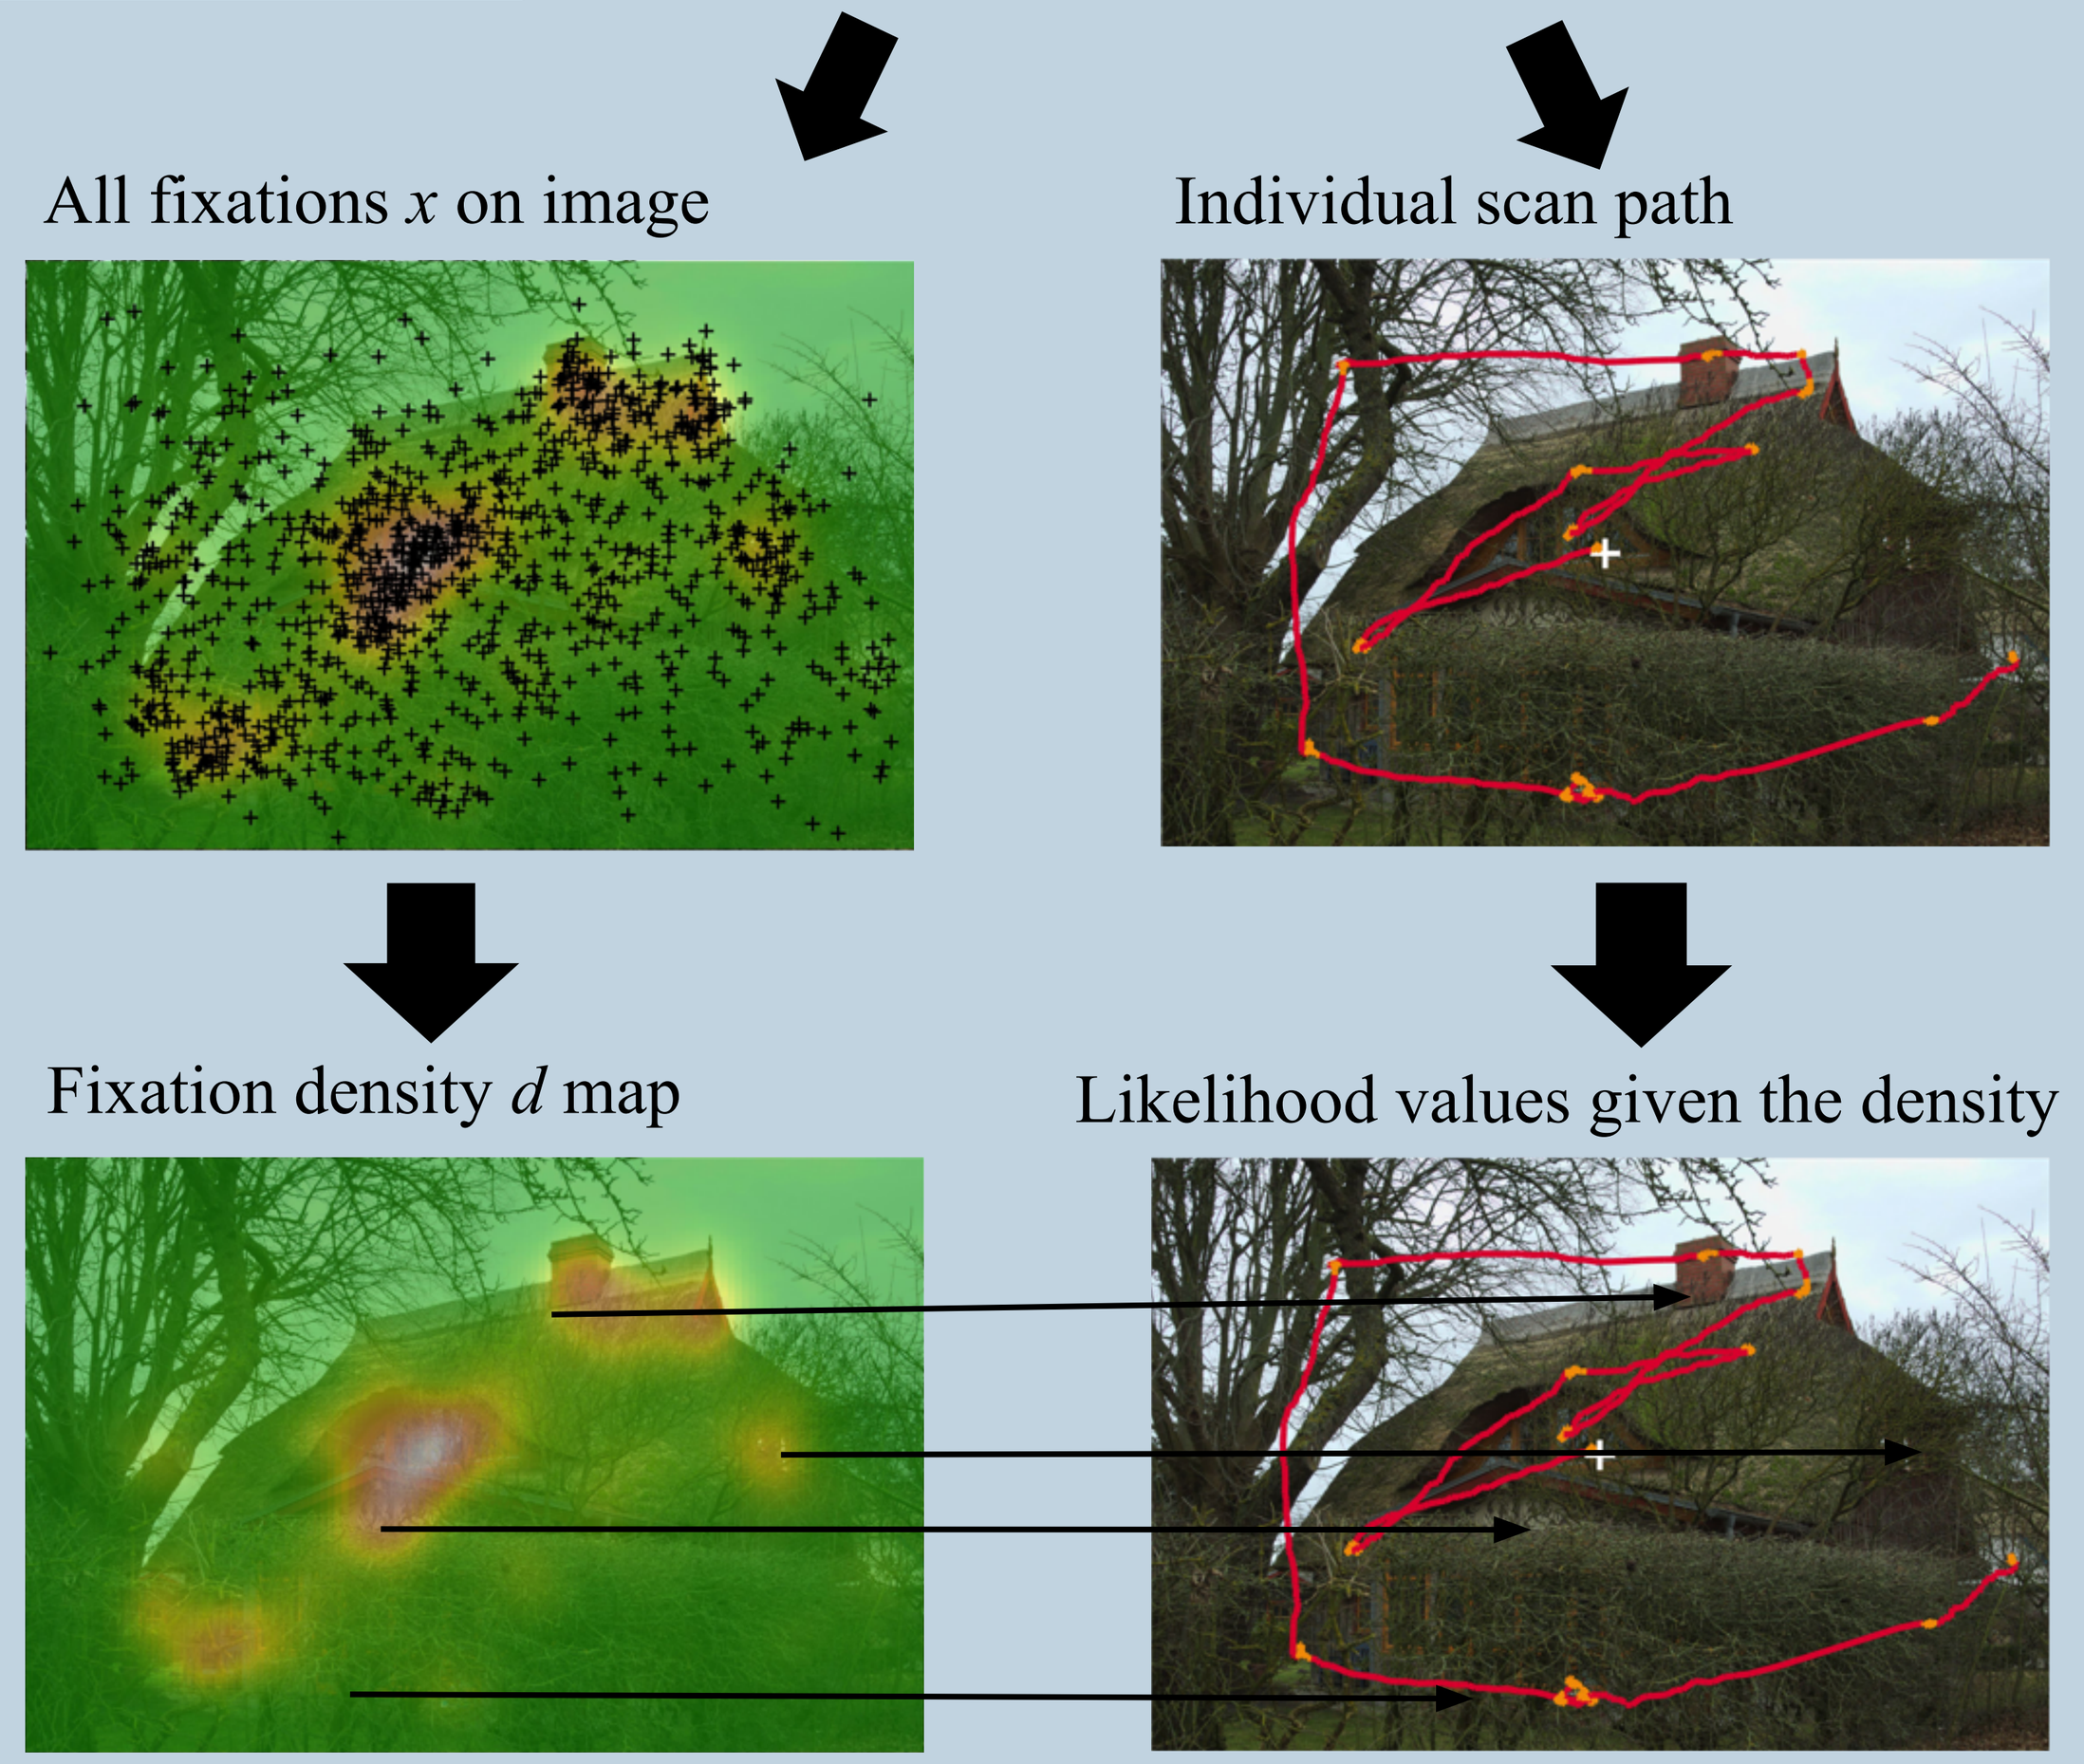

All fixations *x* on image
Individual scan path
Fixation density *d* map
Likelihood values given the density

Conclusions

- Scene exploration is primarily driven by the current visual input and only weakly modulated by VLTM from previous days, or even VLTM from hours and minutes ago
- Even in the absence of peripheral image information we find evidence for only very limited influence of VLTM on eye movement measures
- Possible caveats are the new image material and the introduction of the "similar" condition into the experimental design
- **Take Home Message:** The previously reported effect of familiarity on eye movement seems to be either smaller or more volatile than we expected

References & Aknowledgements

1) Castelhano, M. S., & Henderson, J. M. (2007). Initial scene representations facilitate eye movement guidance in visual search. *Journal of Experimental Psychology: Human Perception and Performance*, 33(4), 753–763.
2) Kaspar, K., & König, P. (2011a). Overt attention and context factors: The impact of repeated presentations, image type, and individual motivation. *PLoS ONE*, 6(7), e21719.
3) Kaspar, K., & König, P. (2011b). Viewing behavior and the impact of low-level image properties across repeated presentations of complex scenes. *Journal of Vision*, 11(13), 26.
4) Trukenbrod, H. A., Barthelmé, S., Wichmann, F. A., & Engbert, R. (accepted). Spatial statistics for gaze patterns in scene viewing: Effects of repeated viewing. *Journal of Vision.*
5) Brady, T. F., Konkle, T., Alvarez, G. A., & Oliva, A. (2008). Visual long-term memory has a massive storage capacity for object details. *Proceedings of the National Academy of Sciences*, 105(38), 14325–14329.

This work was supported by grants from Deutsche Forschungsgemeinschaft to the collaborative research center SFB 1294 project B05

Contact: Lisa.Schwetlick@uni-potsdam.de